Personalized Health Solutions

Device Case Study: myPKFiT for ADVATE / ADYNOVATE

Authors: Jennifer Craig Cordova, Mike Nelson, McCay Barnes, Chad Plautz, Dhairya Mehta, Ken Tubman, and Tim Moloy

Medical Devices Center of Excellences, R&D, Shire HGT, Inc., Lexington, MA, a Takeda company

Introduction: Connected Ecosystem Vision

A Connected eHealth Ecosystem that encompasses the drug, drug delivery, diagnostics, and connectivity and services can enhance the user experience and drive value:

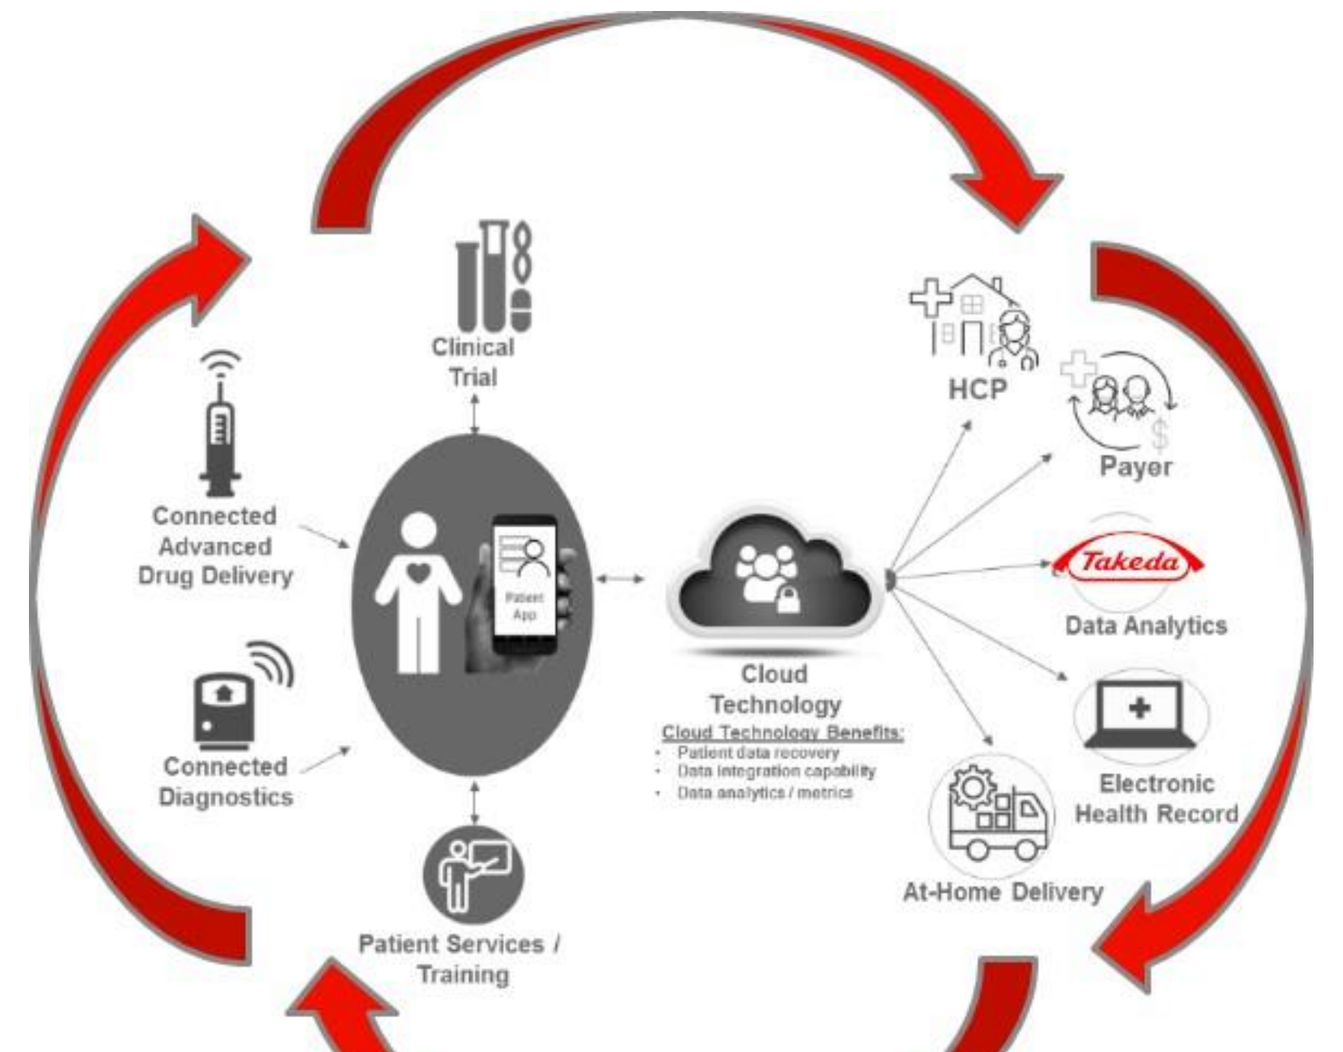
Clinical Trial
HCP
Payor
Connected Advanced Drug Delivery
Data Analytics
Cloud Technology
Cloud Technology Benefits:
Patient data recovery
Data integration capability
Data analytics / metrics
Connected Diagnostics
Electronic Health Record
At-Home Delivery
Patient Services / Training

Example Benefits:

- **Patient Empowerment** - Patients are able to “own treatment” by tracking treatment therapy, owning their data, and sharing data with their healthcare provider.
- **Patient Education** - Patients are able to learn about disease state management and treatment.
- **Drive Adherence** – Patient app tools and education leveraging dosing data (e.g. behavior modification, gamification) can be used to motivate and encourage patient adherence to their HCP’s treatment plan.
- **Generate Healthcare Provider Insights** – HCPs are better informed of their patient’s adherence between office visits, allowing care teams to better track, manage, and adjust patient care plans.
- **Product Feedback Loop** – Takeda learns how patients and HCPs use the connected devices and services in the real world (e.g. which features are used/not used, frequency of use, etc.), which will inform future product features and design improvements.

Case Study: Personalized Health Solution (myPKFiT)

Prophylaxis must be personalized to achieve better outcomes… because everyone is unique. Pharmacokinetics is a critical element of personalization… as interpatient variability creates uncertainty in outcomes.

Unmet Medical Need

- Physicians do not have a way to create a personalized dosing regimen for every patient
- Patients do not have an easy way to keep track of their infusion schedule and bleed activity and share with their care team
- Patients do not have insight to their current FVIII levels

Value Proposition and Differentiation

- Personalized dosing per patient PK curve
- PK dosing based on only 2 blood draws vs 11 previously
- Patients can log their infusion and bleed infusion and easily share with their care team
- Patients can keep track of their FVIII levels in real time following prophylaxis infusions

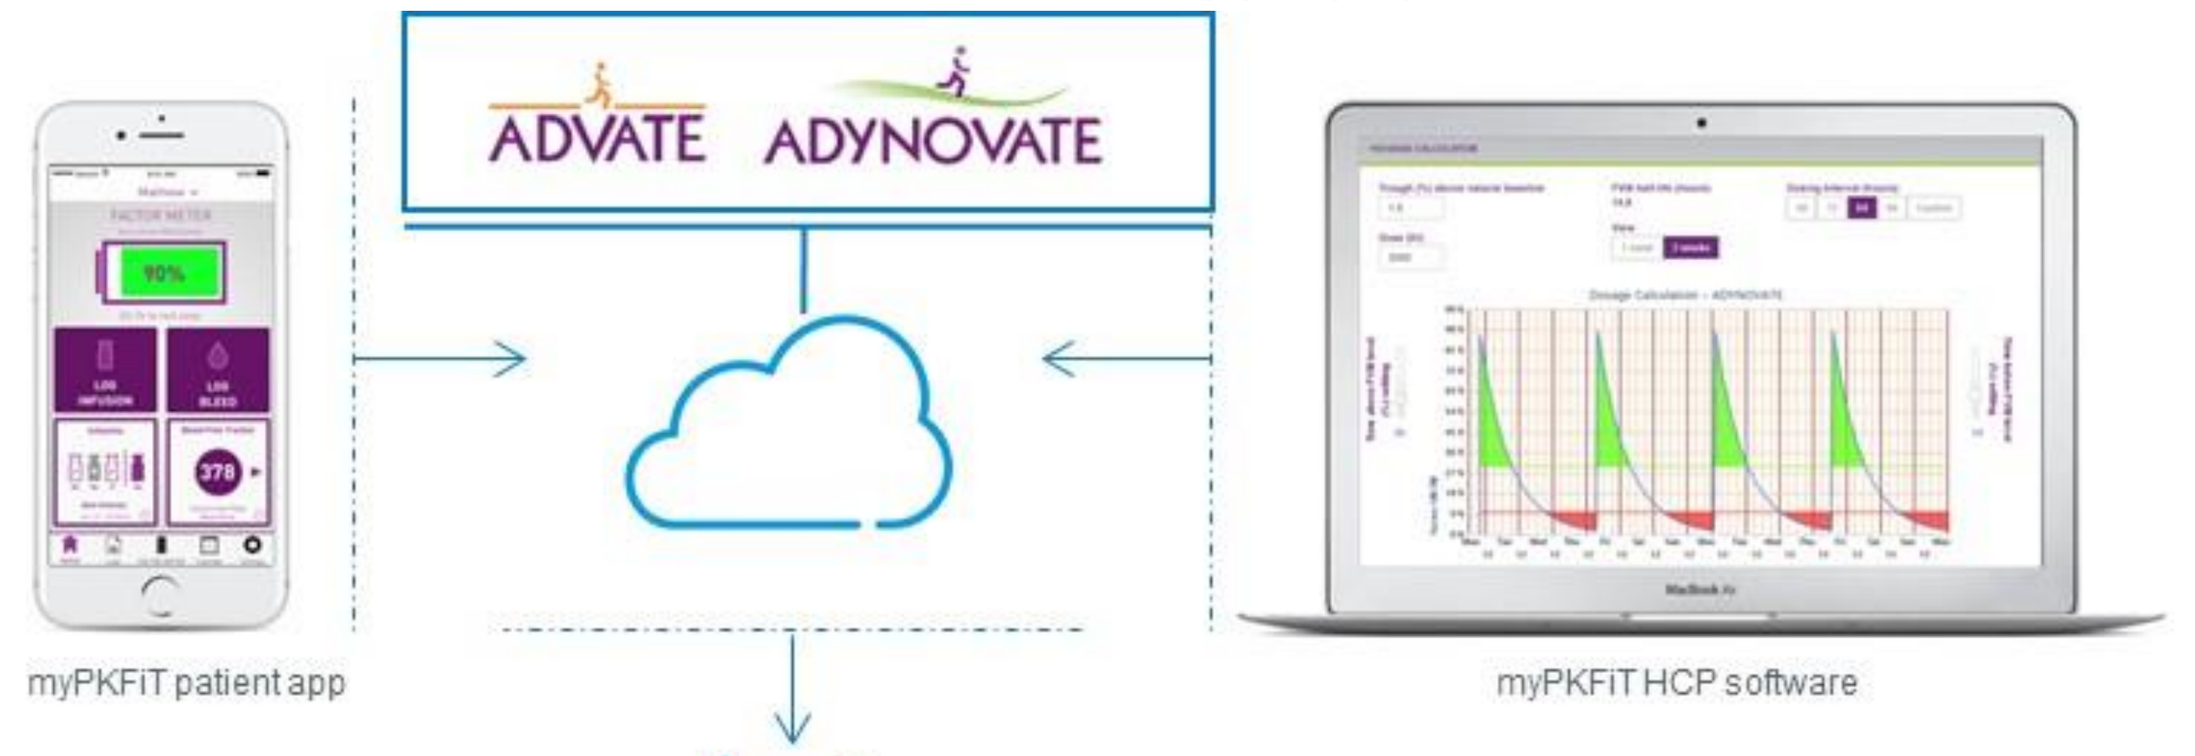
ADVATE
ADYNOVATE
myPKFiT patient app
myPKFiT HCP software
External Data Base

myPKFiT Patient Application (ADYNOVATE / ADVATE)
The myPKFiT Patient APP (cloud) takes the personalized dosing recommendation from the myPKFiT HCP and allows the patient to manage an eDiary (e.g., bleeds and infusions log) and to track factor coverage through the “factor meter”.

myPKFiT HCP v3.0 (ADYNOVATE / ADVATE):
The HCP device is a standalone web-based software medical device intended to be used by healthcare professionals (HCPs) who are providing hemophilia care and provides dosing recommendations.

Solution: myPKFiT: Practical and reliable estimate of individual personalized prophylaxis (PK) regimen

The software output may be used to guide decisions on appropriate dosing and infusion intervals to maintain FVIII activity levels at or above a user specified minimum FVIII activity level between 1% - 3% above natural baseline for an individual patient in accordance with the FDA-approved dosing regimens provided in the ADVATE Prescribing Information (PI).

myPKFiT uses a Bayesian calculation to determine the estimated PK parameters for an individual patient based on population PK model.

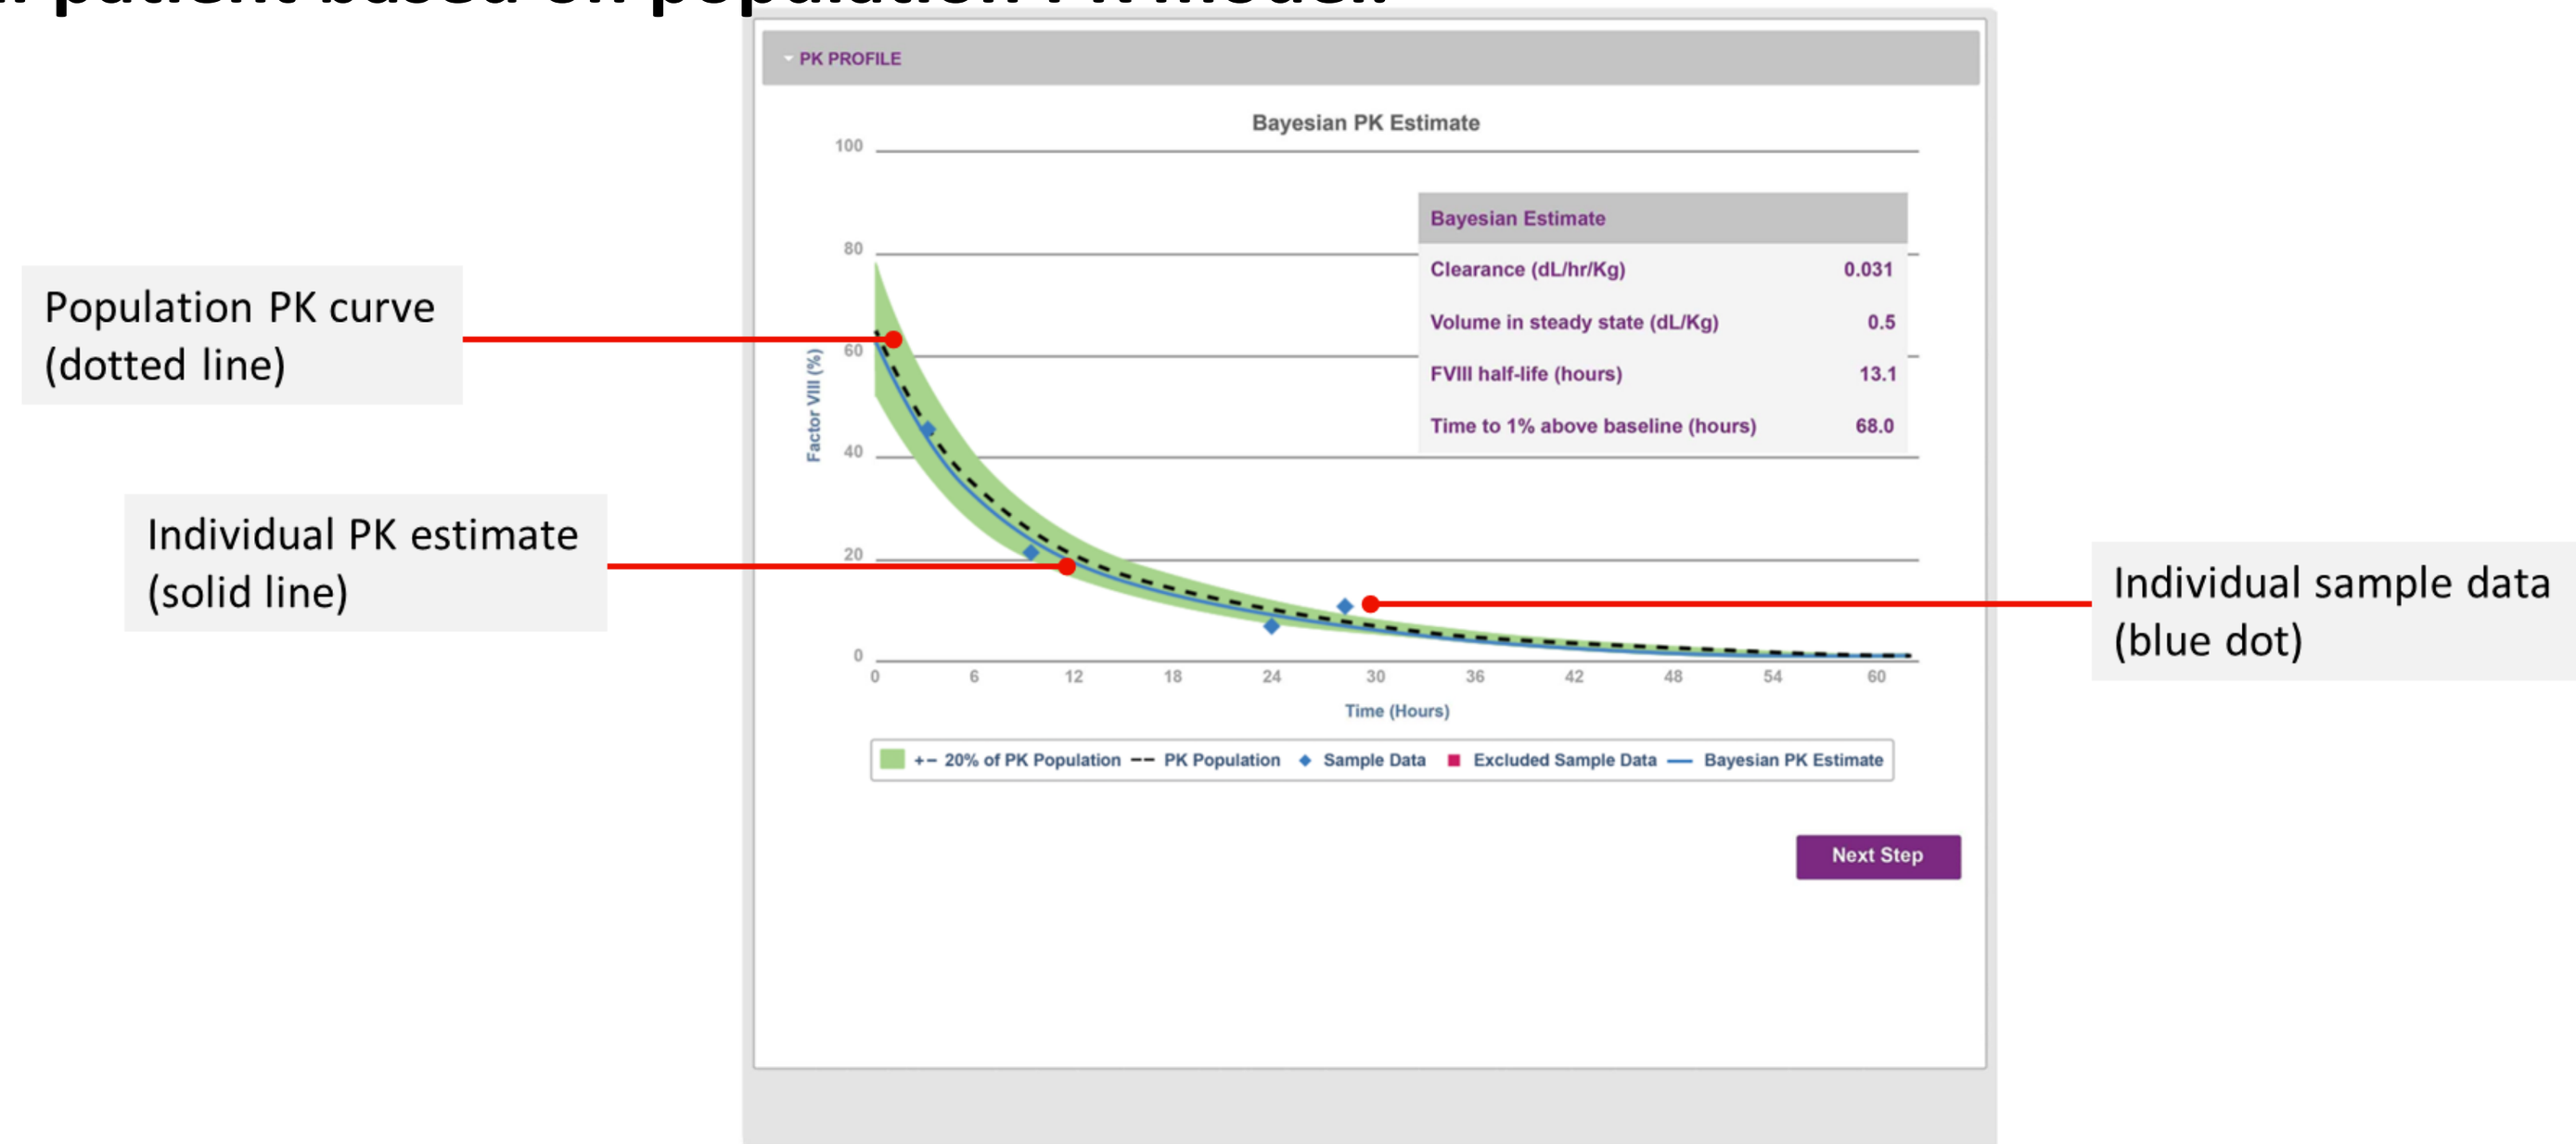
Population PK curve (dotted line)
Individual PK estimate (solid line)
Individual sample data (blue dot)
PK PROFILE
Bayesian PK Estimate
Bayesian Estimate
Clearance (dL/hr/Kg) 0.031
Volume in steady state (dL/Kg) 0.5
FVIII half-life (hours) 13.1
Time to 1% above baseline (hours) 68.0
Factor VIII (%)
Time (Hours)
+– 20% of PK Population
PK Population
Sample Data
Excluded Sample Data
Bayesian PK Estimate
Next Step

myPKFiT allows you to model personalized dosing regimens for your patient based on previously estimated individual PK parameters. Adjustments can be made to visualize target trough above baseline, dosing intervals and assess the dosing schedule in a table format.

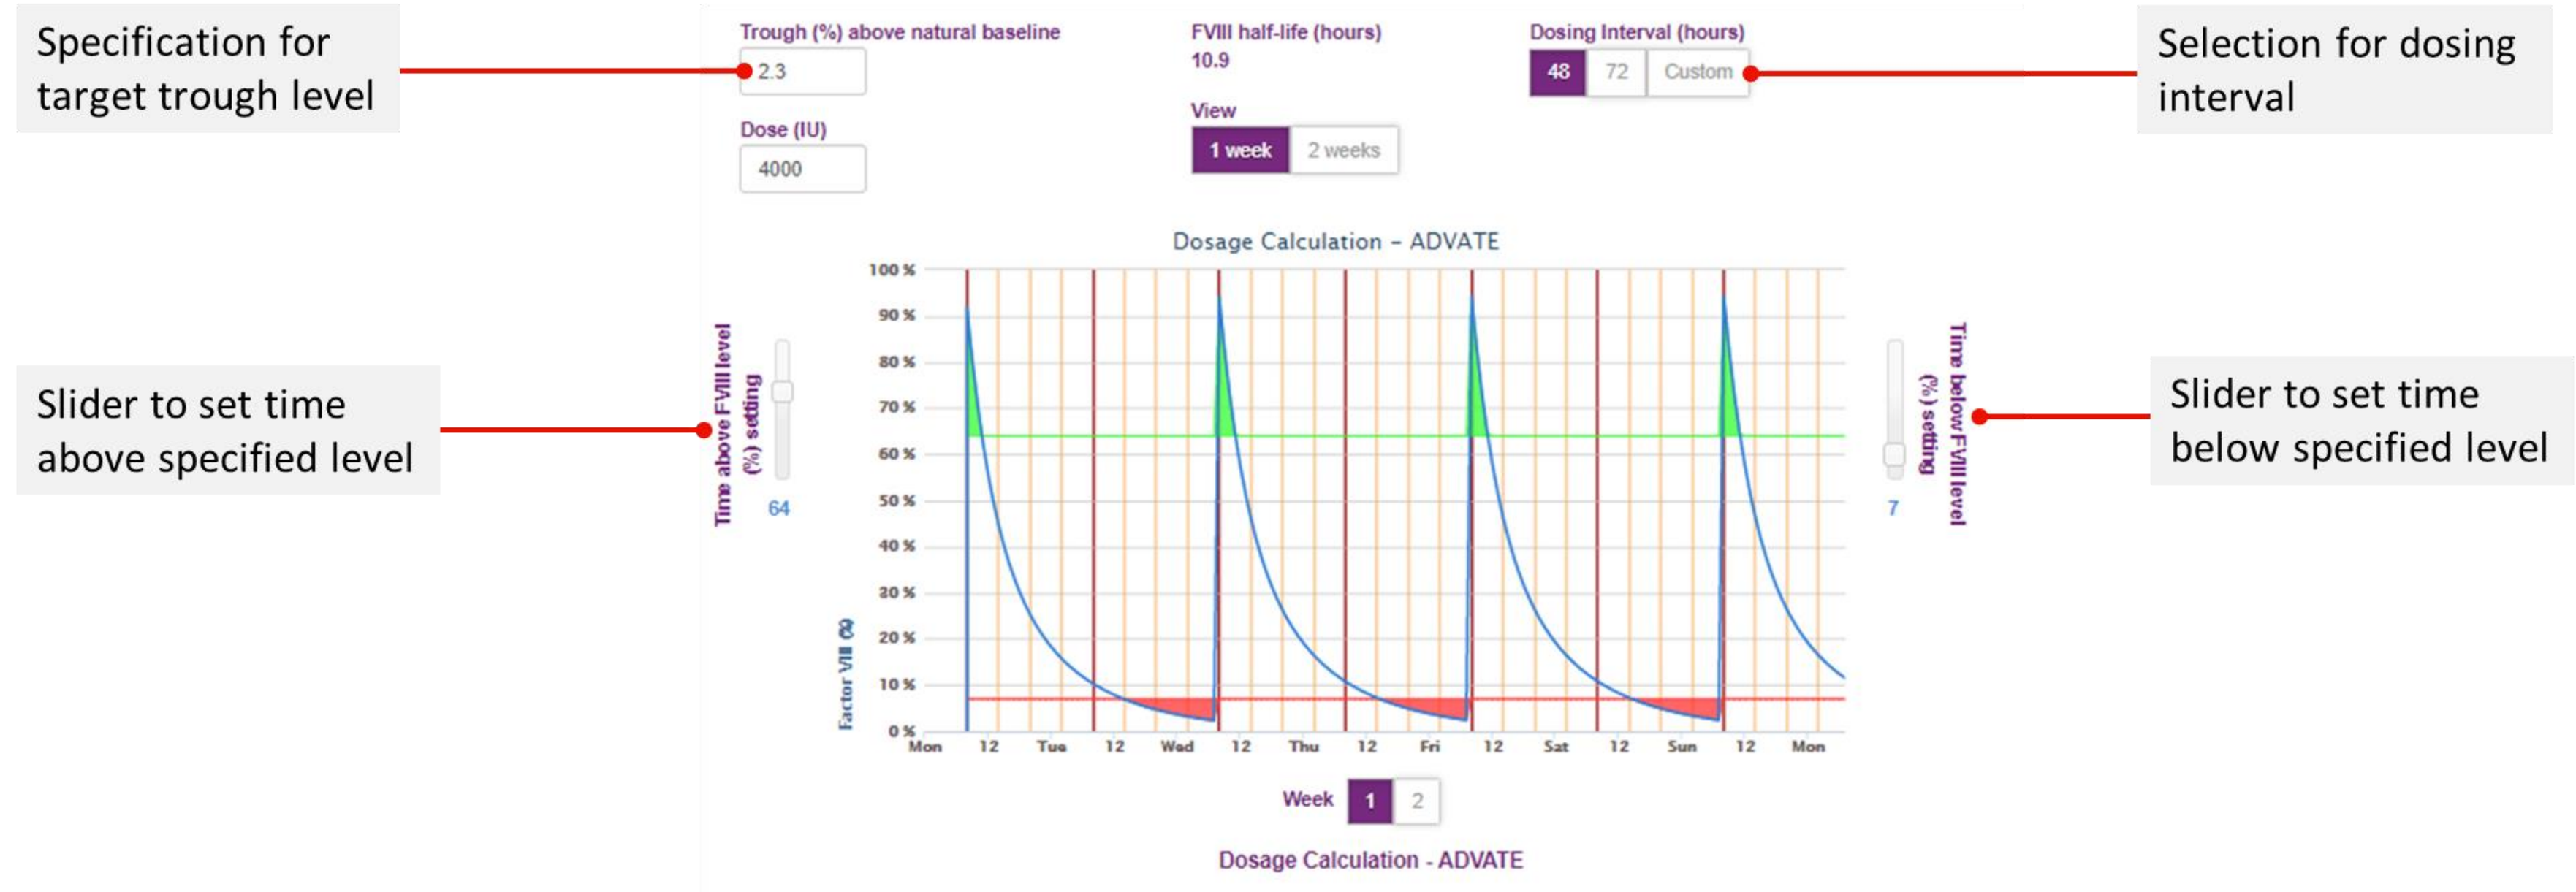
Specification for target trough level
Selection for dosing interval
Slider to set time above specified level
Slider to set time below specified level
Data output table aligned to graph
Trough (%) above natural baseline
2.3
Dose (IU)
4000
FVIII half-life (hours)
10.9
View
1 week
2 weeks
Dosing Interval (hours)
48
72
Custom
Dosage Calculation – ADVATE
Time above FVIII level (%) setting
64
Time below FVIII level (%) setting
7
Factor VIII (%)
Week
Dosage Calculation - ADVATE

Week 1	Mon	Tue	Wed	Thu	Fri	Sat	Sun
Dose IU	4000 IU		4000 IU		4000 IU		4000 IU
Dose IU/kg	40.0 IU/kg		40.0 IU/kg		40.0 IU/kg		40.0 IU/kg
Trough (%) above natural baseline	2.3 %		2.4 %		2.4 %		2.4 %
Time (hrs) above 64%	3 hours		4 hours		4 hours		4 hours
Time (hrs) below 7%	18 hours		17 hours		17 hours		17 hours

Week 2	Mon	Tue	Wed	Thu	Fri	Sat	Sun
Dose IU		4000 IU		4000 IU		4000 IU	
Dose IU/kg		40.0 IU/kg		40.0 IU/kg		40.0 IU/kg	
Trough (%) above natural baseline		2.4 %		2.4 %		2.4 %	
Time (hrs) above 64%		4 hours		4 hours		4 hours	
Time (hrs) below 7%		17 hours		17 hours		17 hours	

The personalized assessment information is saved to a PDF for the patient. The PDF contains a QR code for use in the myPKFiT Mobile Application. The patient gives access to the Healthcare Provider using mobile app. The HCP will then have access to Data Sharing for all consenting patients, patient report, and mobile app usage results.

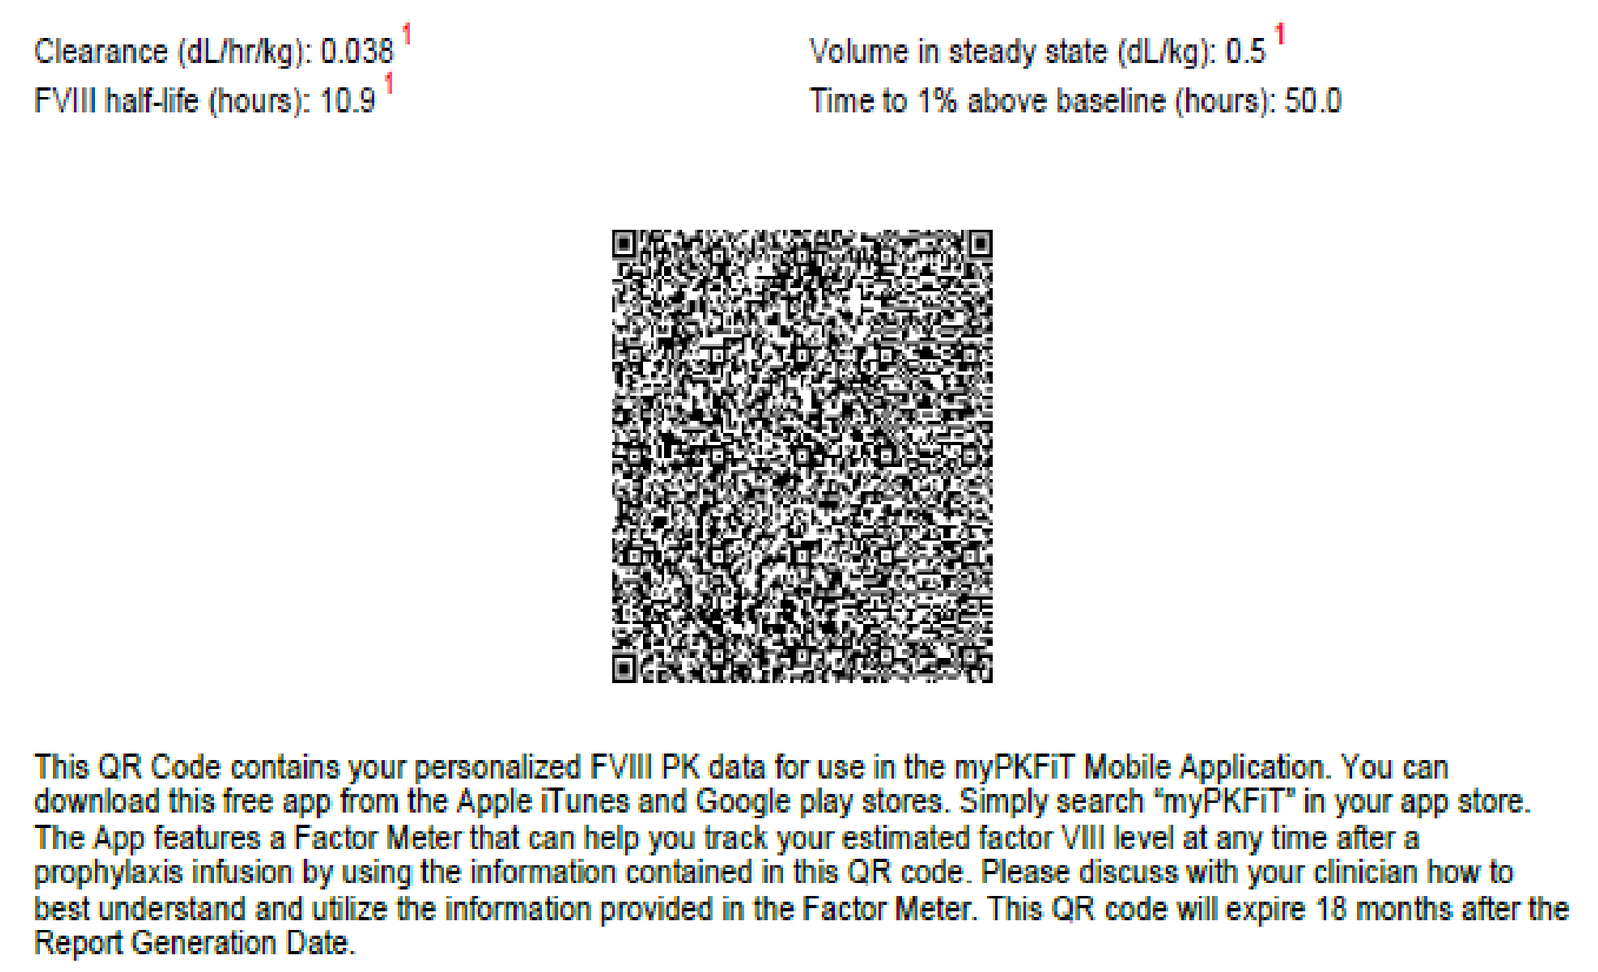
Clearance (dL/hr/kg): 0.038 1
FVIII half-life (hours): 10.9 1
Volume in steady state (dL/kg): 0.5 1
Time to 1% above baseline (hours): 50.0
This QR Code contains your personalized FVIII PK data for use in the myPKFiT Mobile Application. You can download this free app from the Apple iTunes and Google play stores. Simply search “myPKFiT” in your app store. The App features a Factor Meter that can help you track your estimated factor VIII level at any time after a prophylaxis infusion by using the information contained in this QR code. Please discuss with your clinician how to best understand and utilize the information provided in the Factor Meter. This QR code will expire 18 months after the Report Generation Date.

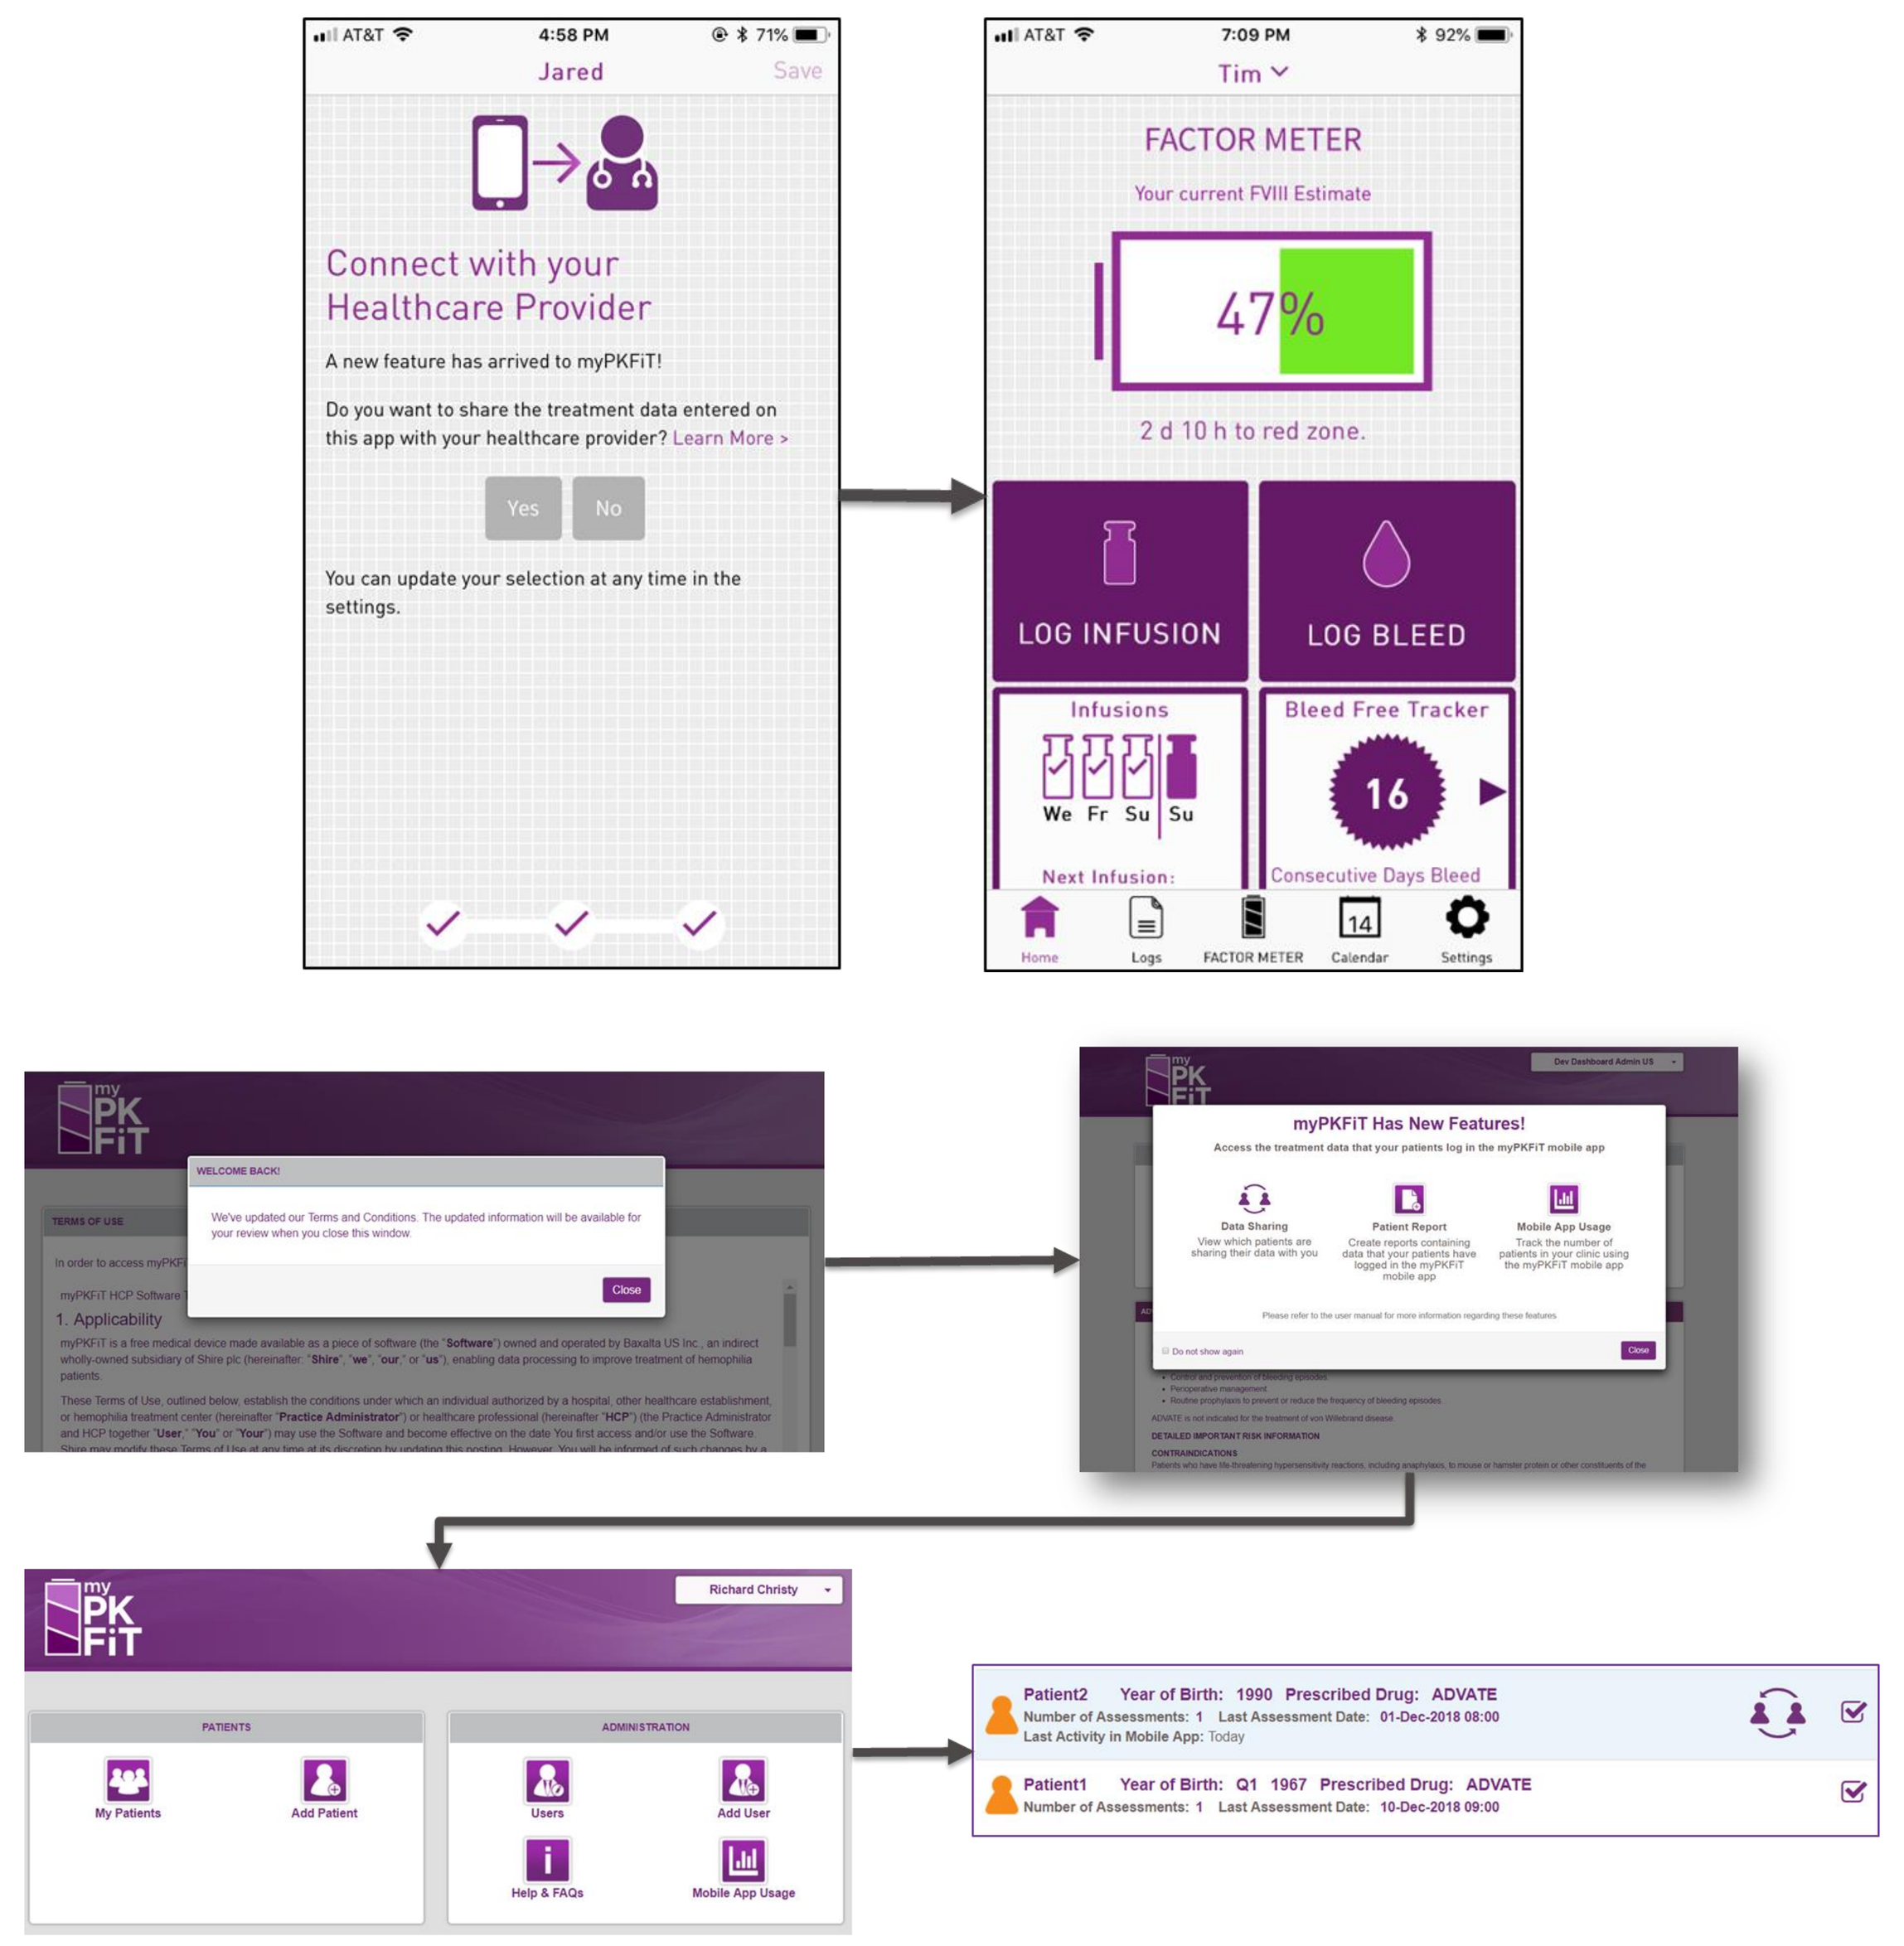
Jared
Save
Connect with your Healthcare Provider
A new feature has arrived to myPKFiT!
Do you want to share the treatment data entered on this app with your healthcare provider? Learn More ›
Yes
No
You can update your selection at any time in the settings.
Tim
FACTOR METER
Your current FVIII Estimate
47%
2 d 10 h to red zone.
LOG INFUSION
LOG BLEED
Infusions
Bleed Free Tracker
16
Consecutive Days Bleed
Next Infusion:
Home
Logs
FACTOR METER
Calendar
Settings
myPKFiT Has New Features!
Data Sharing
Patient Report
Mobile App Usage
WELCOME BACK!
1. Applicability
Close
PATIENTS
ADMINISTRATION
My Patients
Add Patient
Users
Add User
Help & FAQs
Mobile App Usage
Richard Christy
Patient2 Year of Birth: 1990 Prescribed Drug: ADVATE
Patient1 Year of Birth: Q1 1967 Prescribed Drug: ADVATE

Looking Forward: Personalization Health Solutions

The Connected Ecosystem is foundational to Personalized Health Solutions. Increasingly, connected and software devices will utilize data to:

- Gain insights
- Improve our products
- Provide seamless data share within the care network
- Improve research
- Drive decisions
- Demonstrate outcomes

Consideration: Emerging Compliance Environment

Device classification is dependent on geography of WHERE the device is used and HOW the device is used. At summary level, device functionality includes although is not limited to:

- ✓ **Software performs an action on data**
- ✓ **Software performs a measuring function**
- ✓ **Software data is used by Health Care Professional to make decisions**

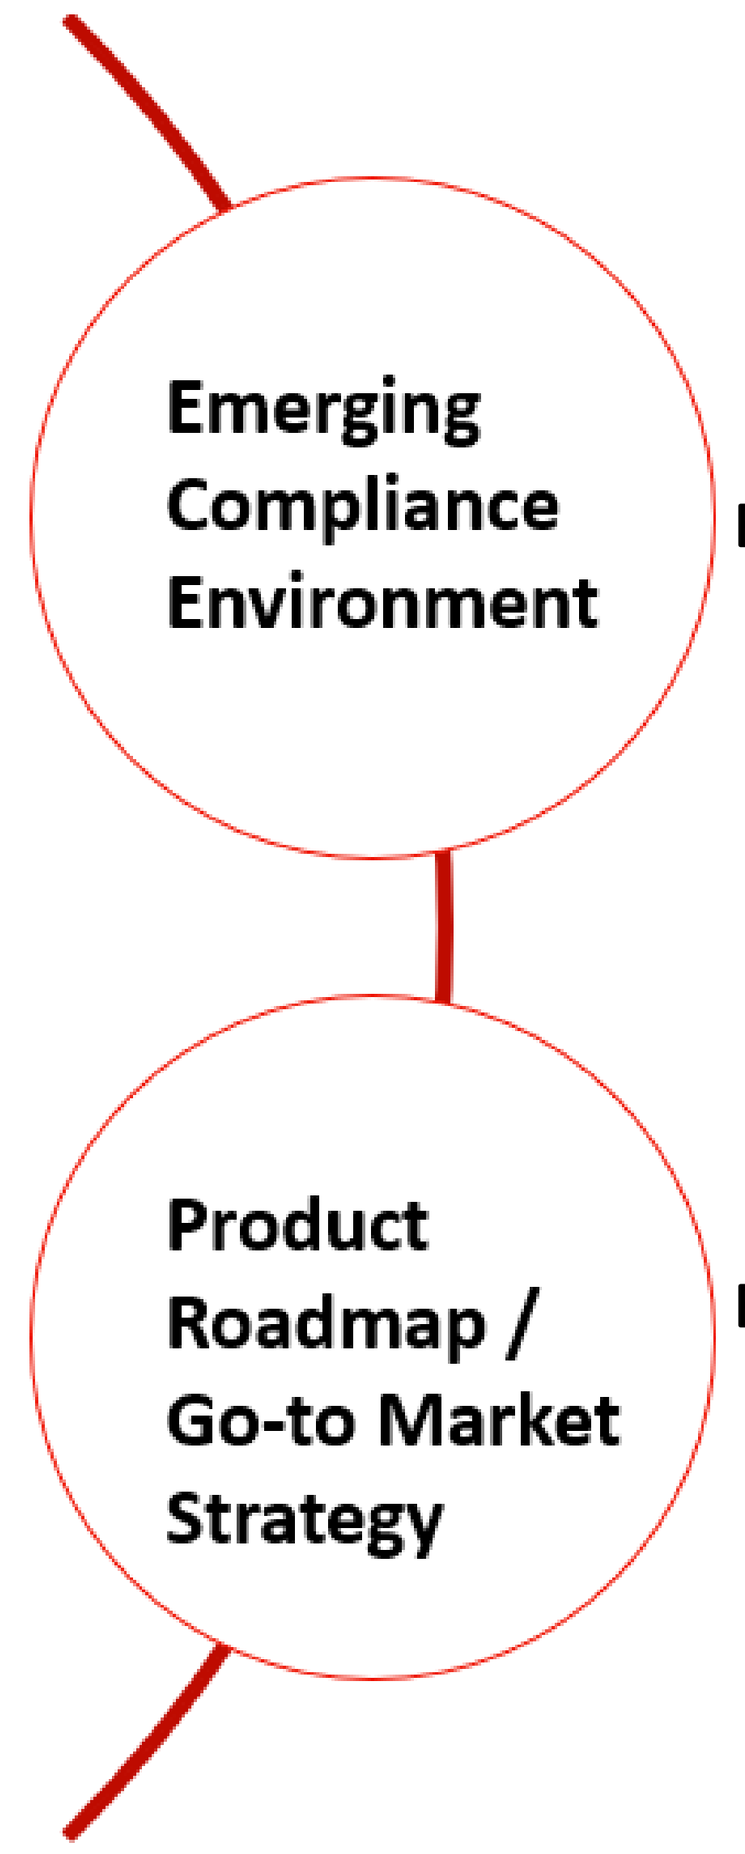
Emerging Compliance Environment
Product Roadmap / Go-to Market Strategy

Examples:

- Unique County Hosting requirements: France (ASIP – Santé); Russia, China
- Unique and changing qualifications: QMS qualifications include: ISO13485, IEC 62304, ISO14971, MDSAP, MDR, etc.

Examples:

- Product Feature Expansion / Geo-Graphic Expansion: As connected platforms are used and features expanded (including drug product references), then the intended use of devices on market may change and may require regulatory updates; or, device classification may be required.

Technical Disclosures

- The myPKFiT Device has been developed by internal cross-functional Takeda device teams (e.g., Medical Devices CoE, R&D; Global Product Teams Commercial; Global Medical Affairs; Global Device Regulatory; Global Device Quality Management; Legal; Data Privacy; etc.) and funded internally by Takeda. myPKFiT has several international patents pending. The myPKFiT personalization is available only for the Takeda products ADVATE and ADYNOVATE. myPKFiT for ADVATE and myPKFiT for ADYNOVATE are commercially launched devices following device design controls in compliance with ISO:13485/MDSAP guidance. The myPKFiT device label is noted as being manufactured by “Shire plc, a wholly owned subsidiary of Takeda”.
- The screen shots are taken with mock patient data from a demonstration (non-production) environment although reflective of the production myPKFiT version. No personal health data was used to create this poster.
- The future of personalized health solutions are the opinions of the Takeda Medical Devices CoE.

PRIMSE Forum Technical Fall Meeting 2019 / https://www.prismeforum.org/
Date: 21st of November, 2019

Takeda Pharmaceutical Company Limited