Nephro-oncology: Closer monitoring of renal function in early-phase trials

Laura Hutchinson[1], Paul Fitzpatrick[2], Jenny Royle[1], Leanne Ogden[1,3], Dónal Landers[1,4]
[1]digital Experimental Cancer Medicine Team, Cancer Research UK Manchester Institute, [2]PF Data Innovations, [3]Manchester University NHS Foundation Trust, [4]AstraZeneca

Digital Experimental Cancer Medicine Team
digitalecmt.org

Challenging current eligibility criteria

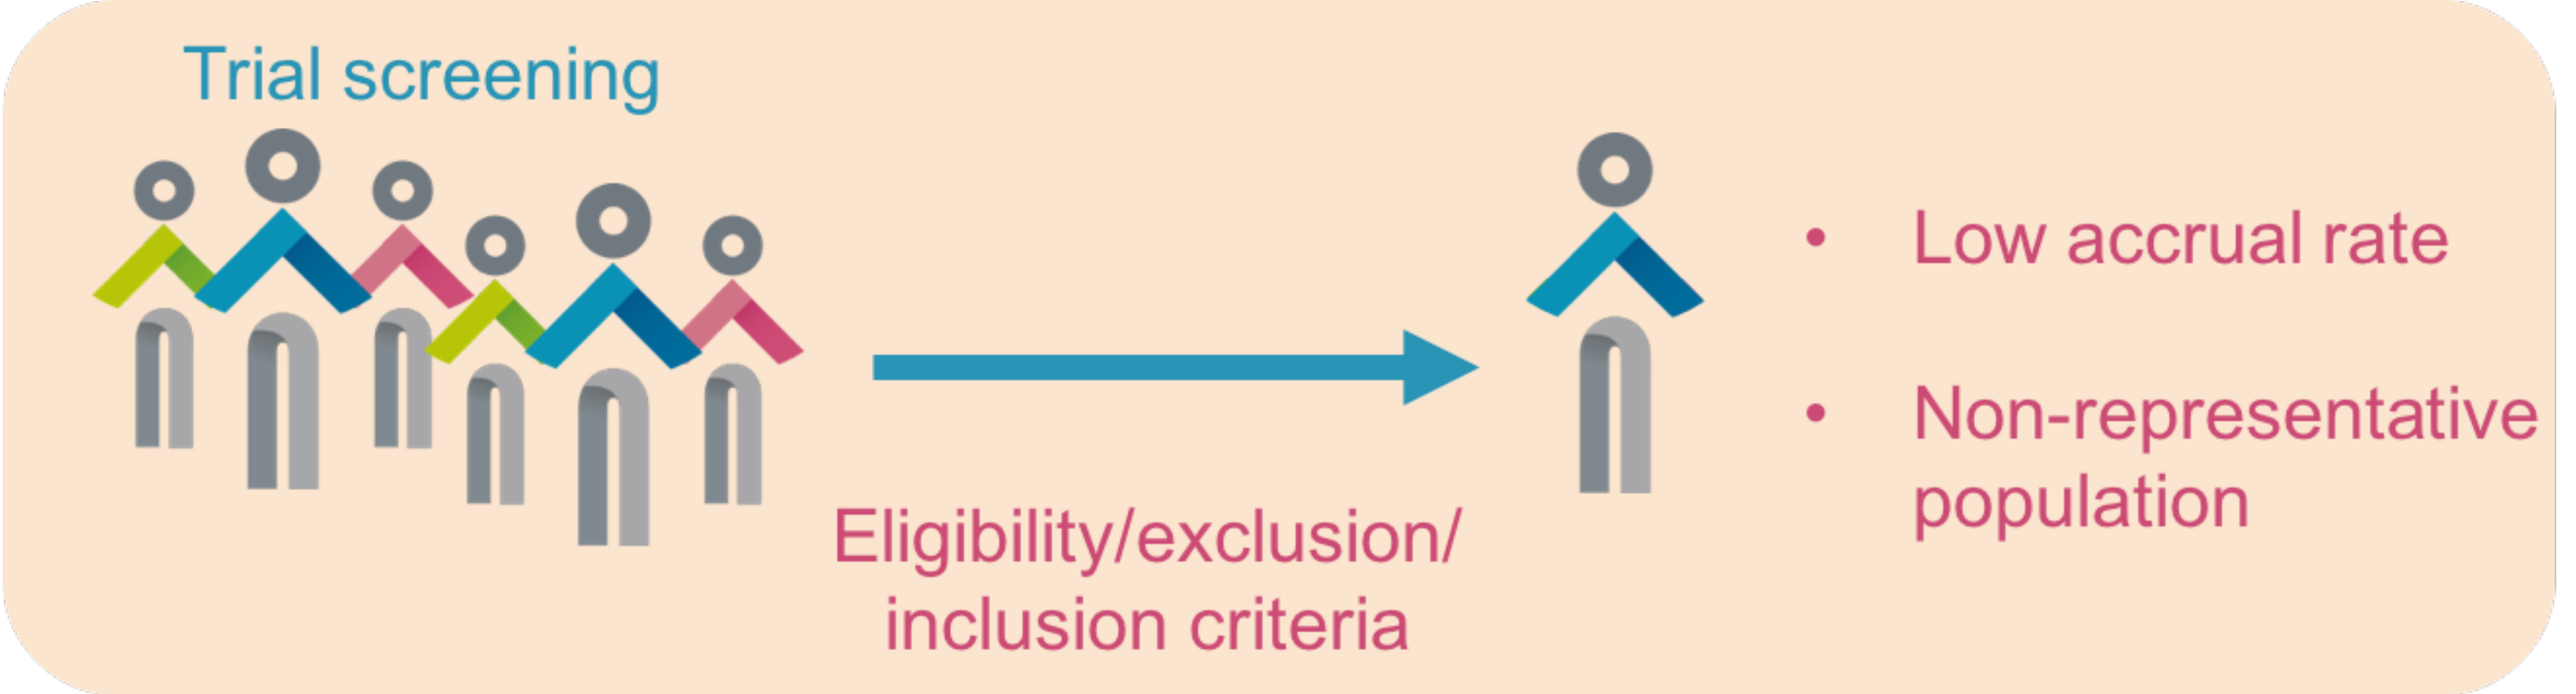
Trial screening
Eligibility/exclusion/ inclusion criteria
Low accrual rate
Non-representative population

Common eligibility criteria for renal function: >50ml/min eGFR

Objective

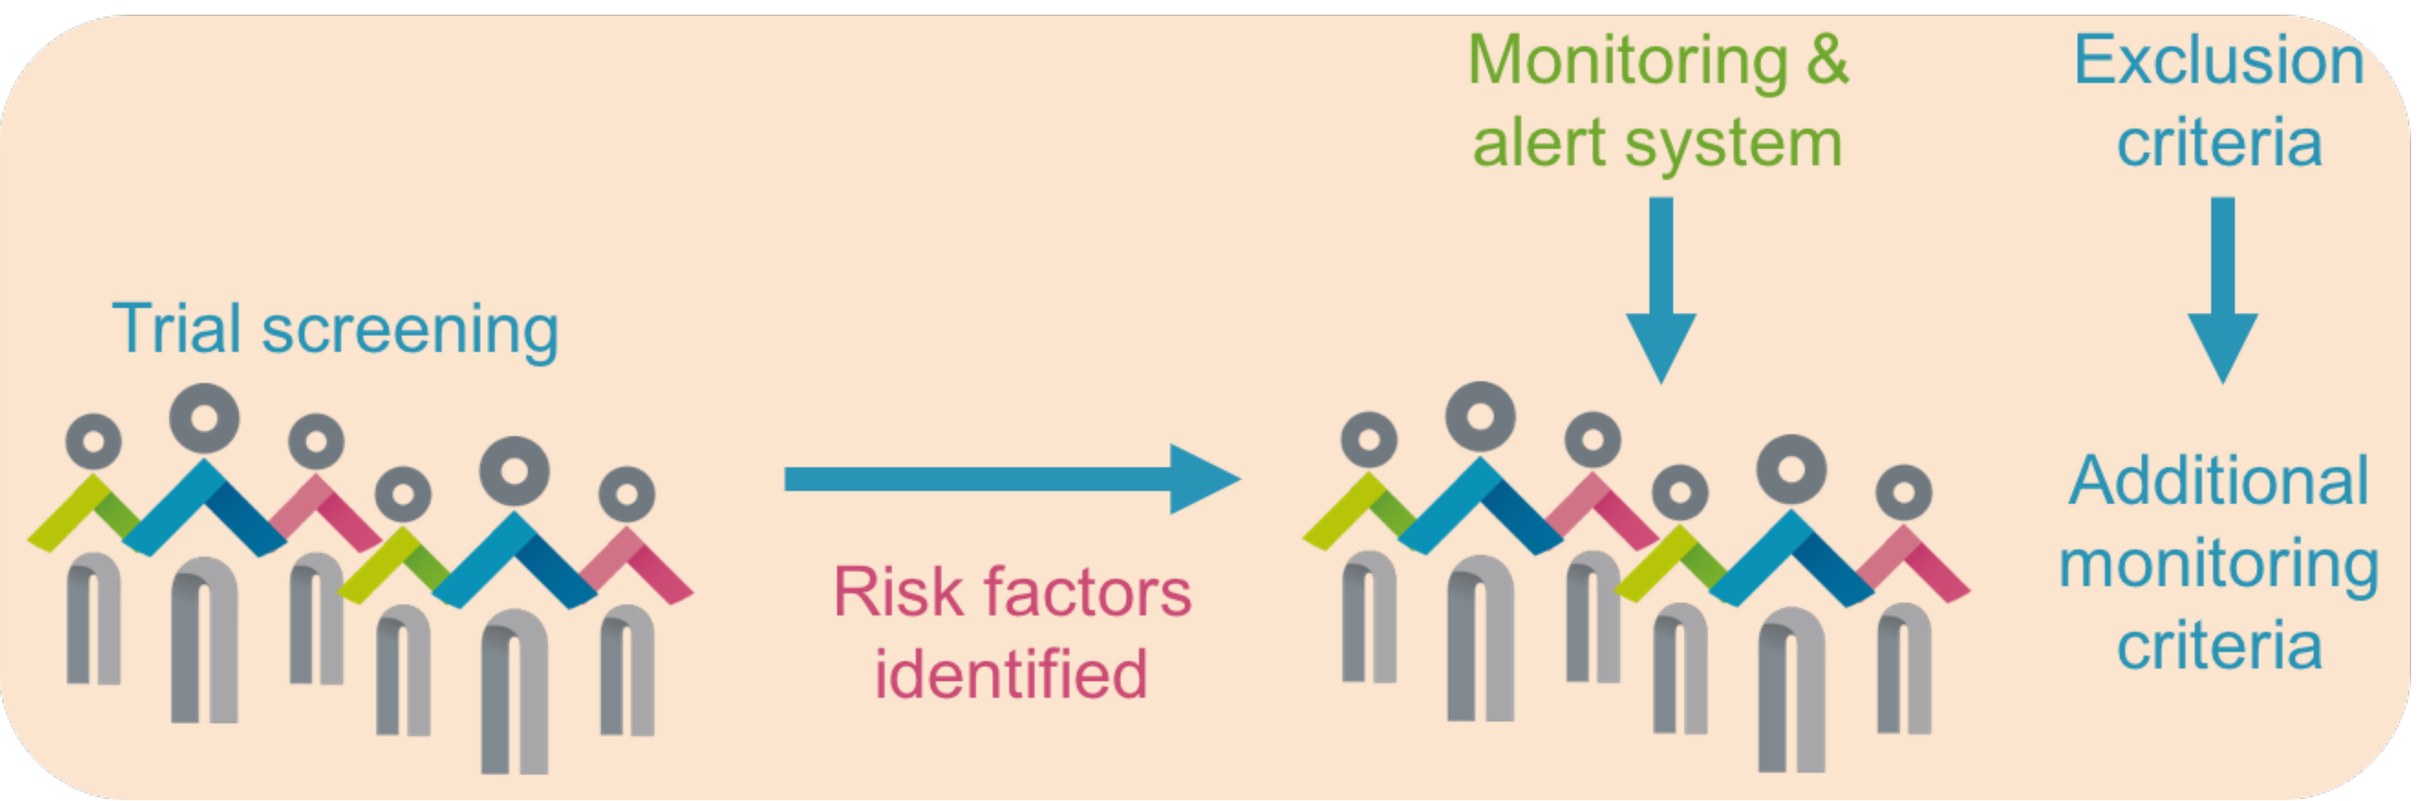
Trial screening
Risk factors identified
Monitoring & alert system
Exclusion criteria
Additional monitoring criteria

To determine if patients with controlled renal dysfunction can participate in early clinical trials

Approach

1 Assess the feasibility of repurposing an NHS approved algorithm for risk classification of acute kidney injury in oncology clinical trials

2 Develop a system that closely monitors risk of acute kidney injury occurring in subjects on an oncology clinical trial

Algorithm

- NHS England endorsed algorithm (NHS England patient safety alert: NHS/PSA/D/2014/010)
- Intended to ensure a timely and consistent approach to the detection of Acute Kidney Injury (AKI)
- Based on serum creatinine changes with time
- Implemented using Microsoft LogicApps
- Triggered by a new measurement
- Processes readings and able to send alerts in real-time

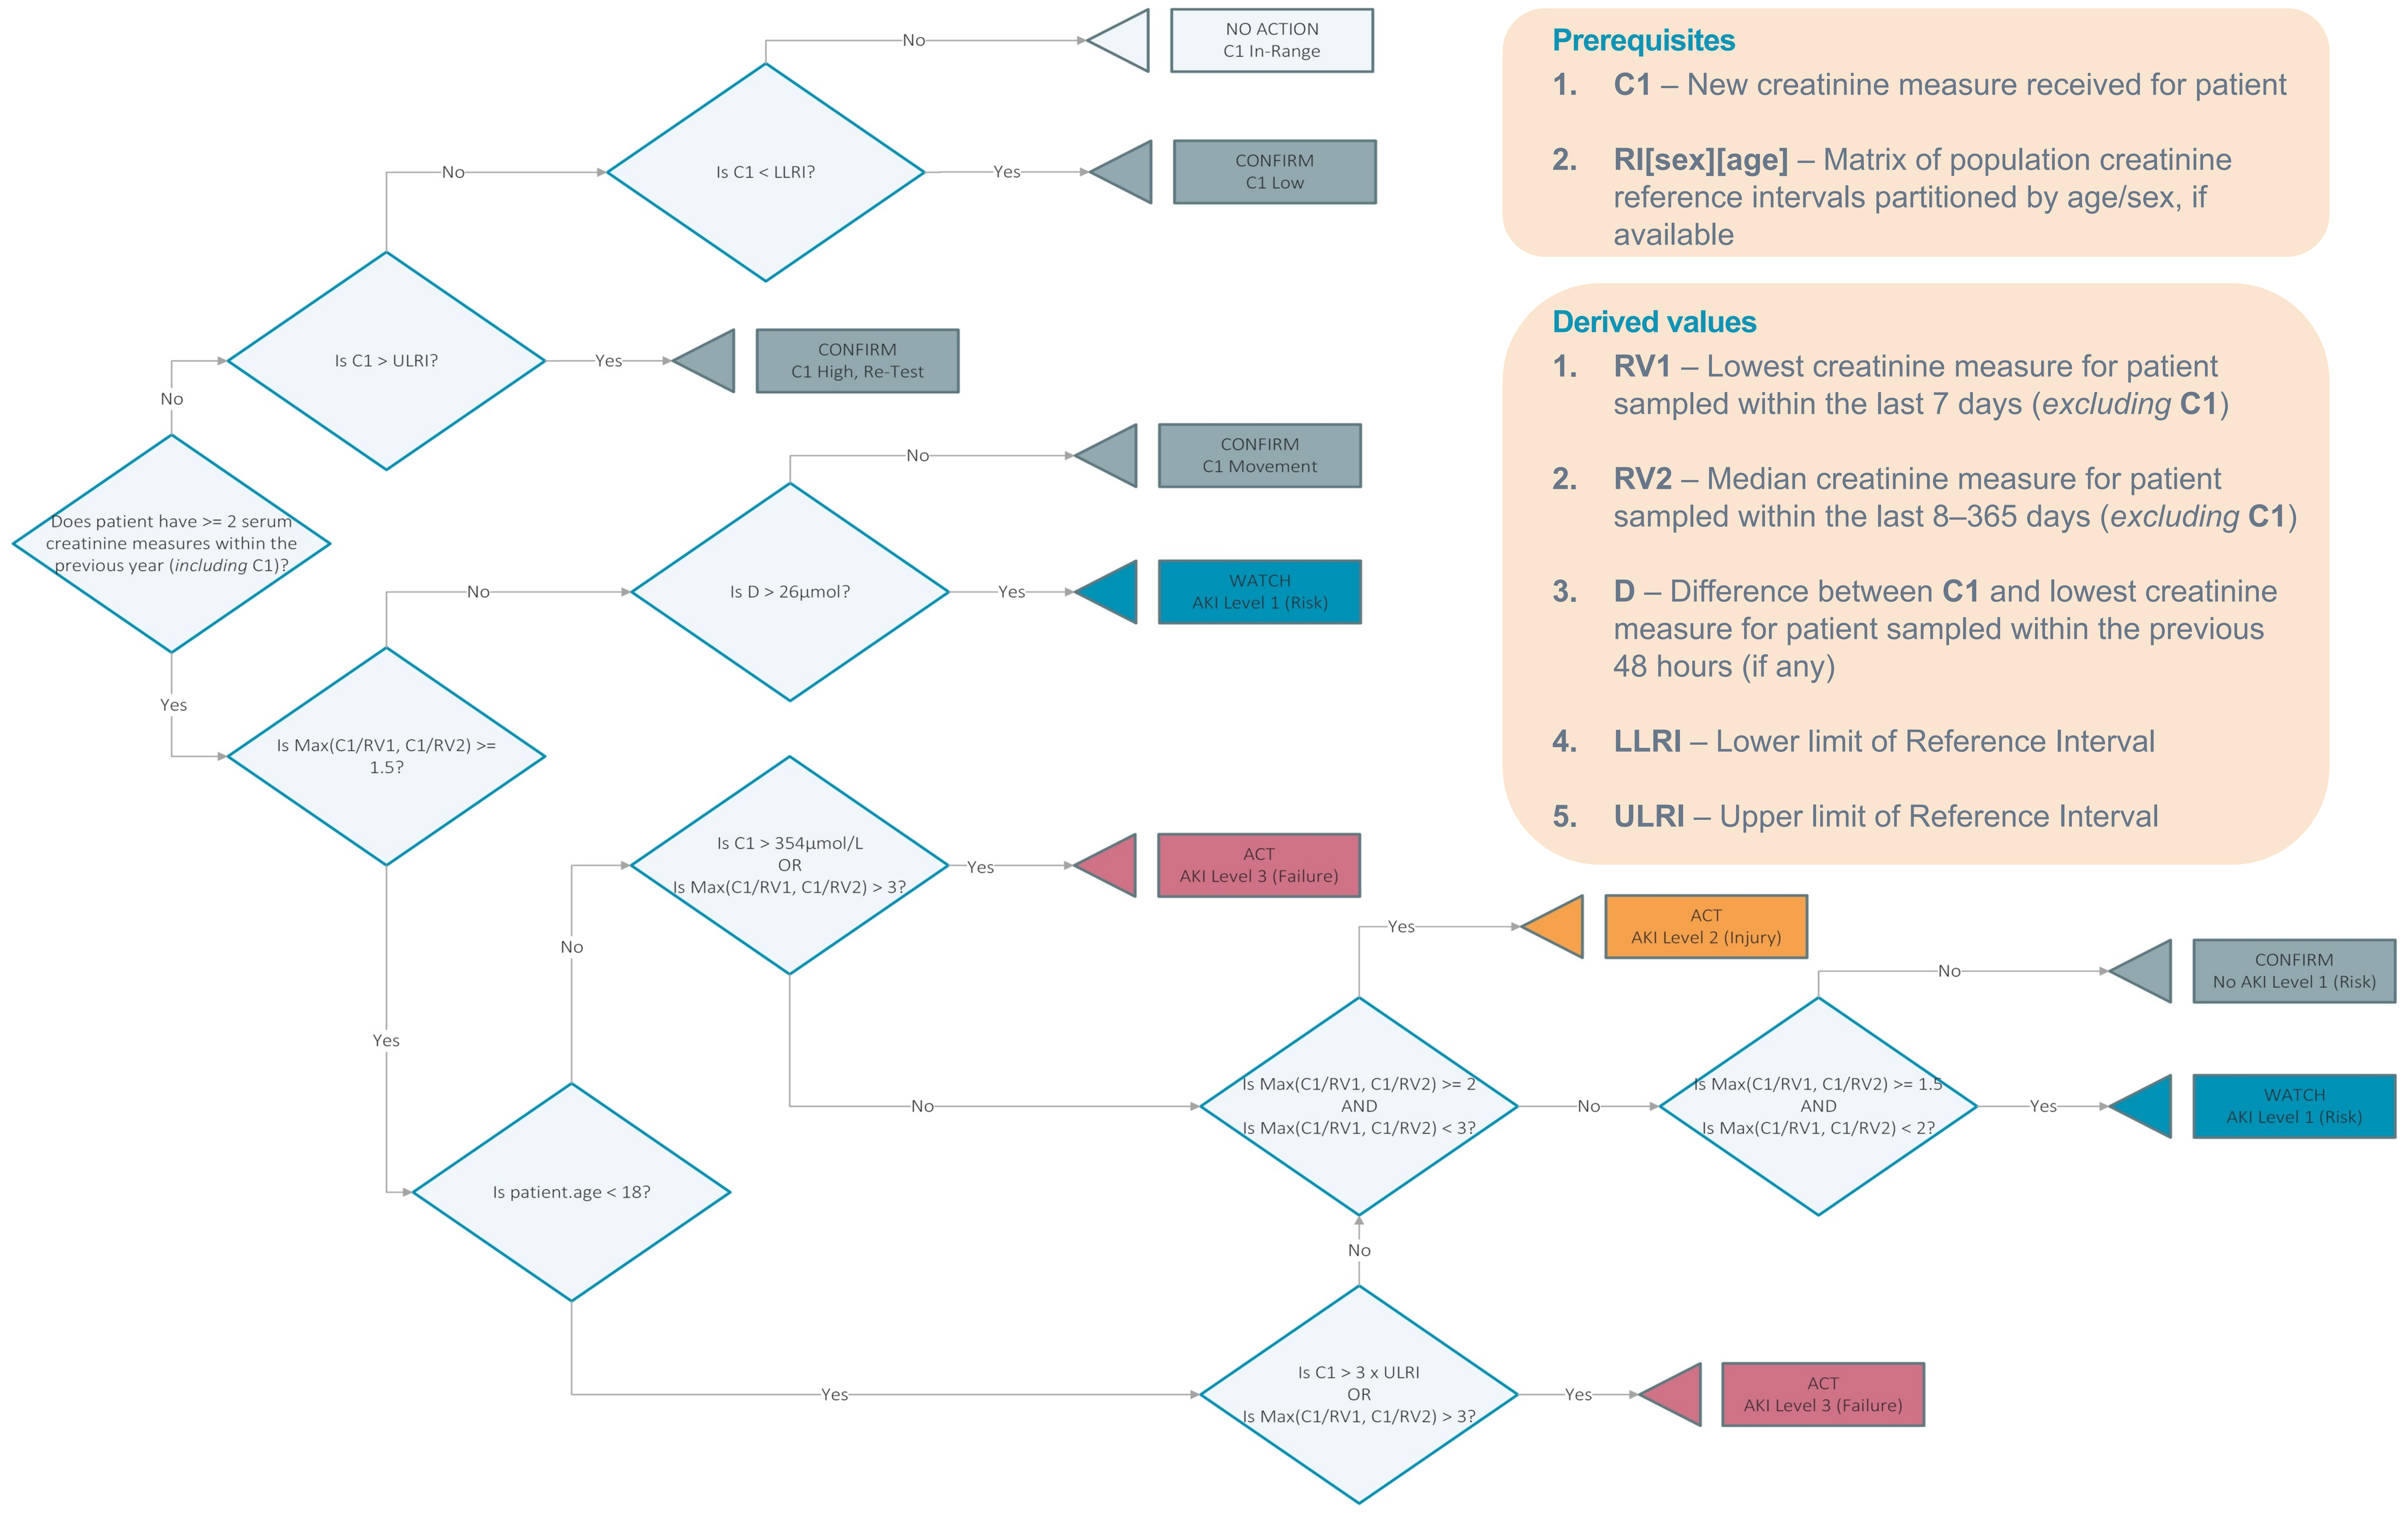
Does patient have >= 2 serum creatinine measures within the previous year (including C1)?
Is C1 > ULRI?
Is C1 < LLRI?
NO ACTION
C1 In-Range
CONFIRM
C1 Low
CONFIRM
C1 High, Re-Test
Is Max(C1/RV1, C1/RV2) >= 1.5?
Is D > 26µmol?
CONFIRM
C1 Movement
WATCH
AKI Level 1 (Risk)
Is C1 > 354µmol/L OR Is Max(C1/RV1, C1/RV2) > 3?
ACT
AKI Level 3 (Failure)
Is patient.age < 18?
Is Max(C1/RV1, C1/RV2) >= 2 AND Is Max(C1/RV1, C1/RV2) < 3?
ACT
AKI Level 2 (Injury)
Is Max(C1/RV1, C1/RV2) >= 1.5 AND Is Max(C1/RV1, C1/RV2) < 2?
CONFIRM
No AKI Level 1 (Risk)
WATCH
AKI Level 1 (Risk)
Is C1 > 3 x ULRI OR Is Max(C1/RV1, C1/RV2) > 3?
ACT
AKI Level 3 (Failure)
Yes
No

Prerequisites

1. **C1** – New creatinine measure received for patient
2. **RI[sex][age]** – Matrix of population creatinine reference intervals partitioned by age/sex, if available

Derived values

1. **RV1** – Lowest creatinine measure for patient sampled within the last 7 days (*excluding* **C1**)
2. **RV2** – Median creatinine measure for patient sampled within the last 8–365 days (*excluding* **C1**)
3. **D** – Difference between **C1** and lowest creatinine measure for patient sampled within the previous 48 hours (if any)
4. **LLRI** – Lower limit of Reference Interval
5. **ULRI** – Upper limit of Reference Interval

Device & applications

Point of Care Creatinine device
Nova® Biomedical StatSensor® Xpress™

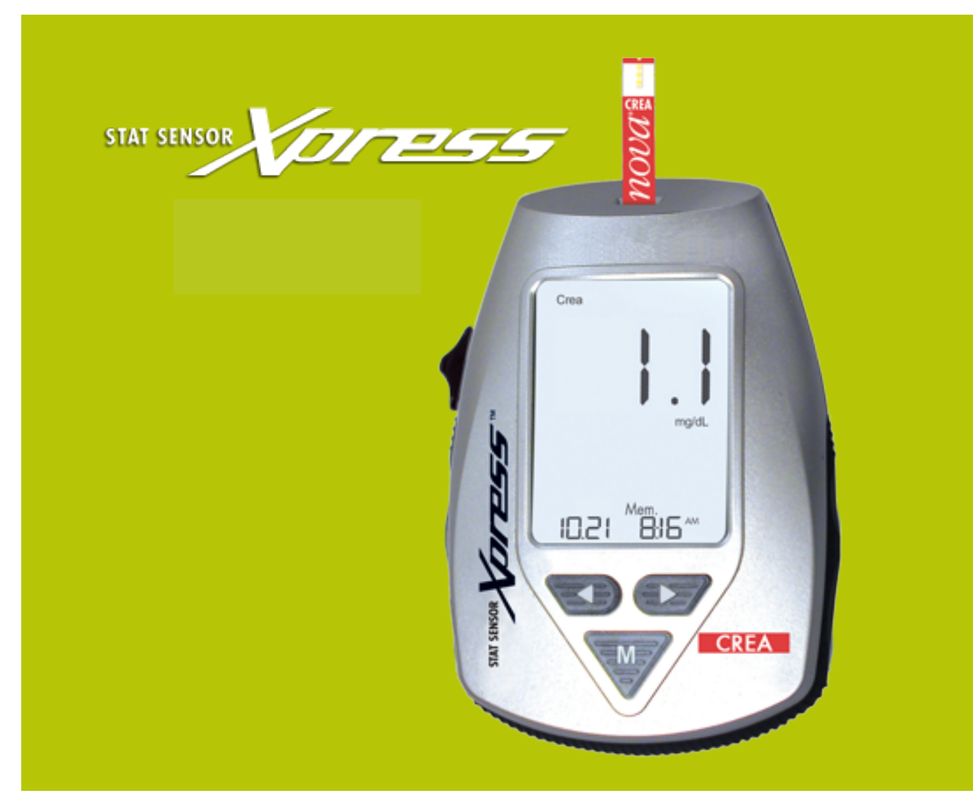
STAT SENSOR Xpress
nova
CREA

- Finger stick capillary blood sampling
- Result in 30 seconds
- Large LCD display
- Limited connectivity (strip port connection to USB; Nova data transfer software)

Patient app
Microsoft PowerApps proof of concept

- Simple UI for uploading measurement and photo of device display
- Mock-ups for displaying patient data have been evaluated at focus groups

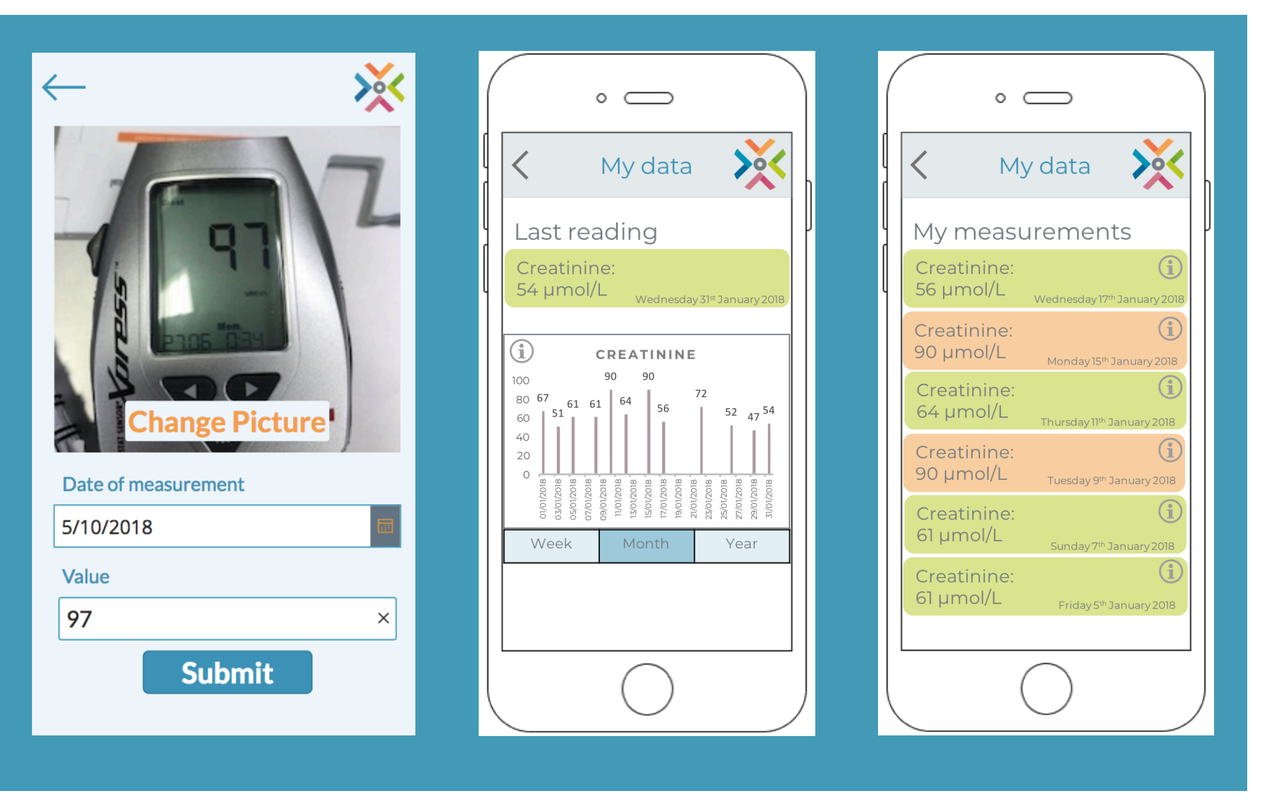
Change Picture
Date of measurement
5/10/2018
Value
97
Submit
My data
Last reading
CREATININE
Week
Month
Year
My data
My measurements

Clinician app
Power BI & Digital Analytics Centre

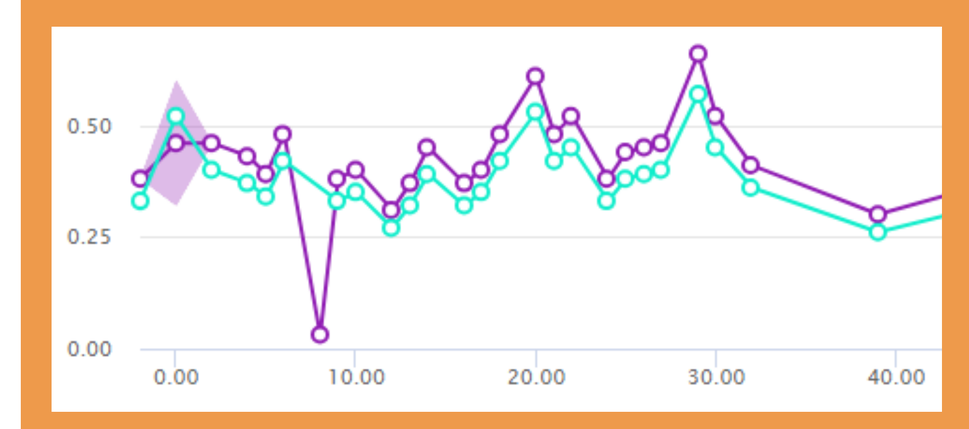
0.50
0.25
0.00
0.00
10.00
20.00
30.00
40.00

- Combining Power BI as a 'first look' and Digital Analytics Centre for more in-depth analysis
- Patient data are displayed within labs line plots
- Clinician app will be designed with input from nephrologists and oncologists

System

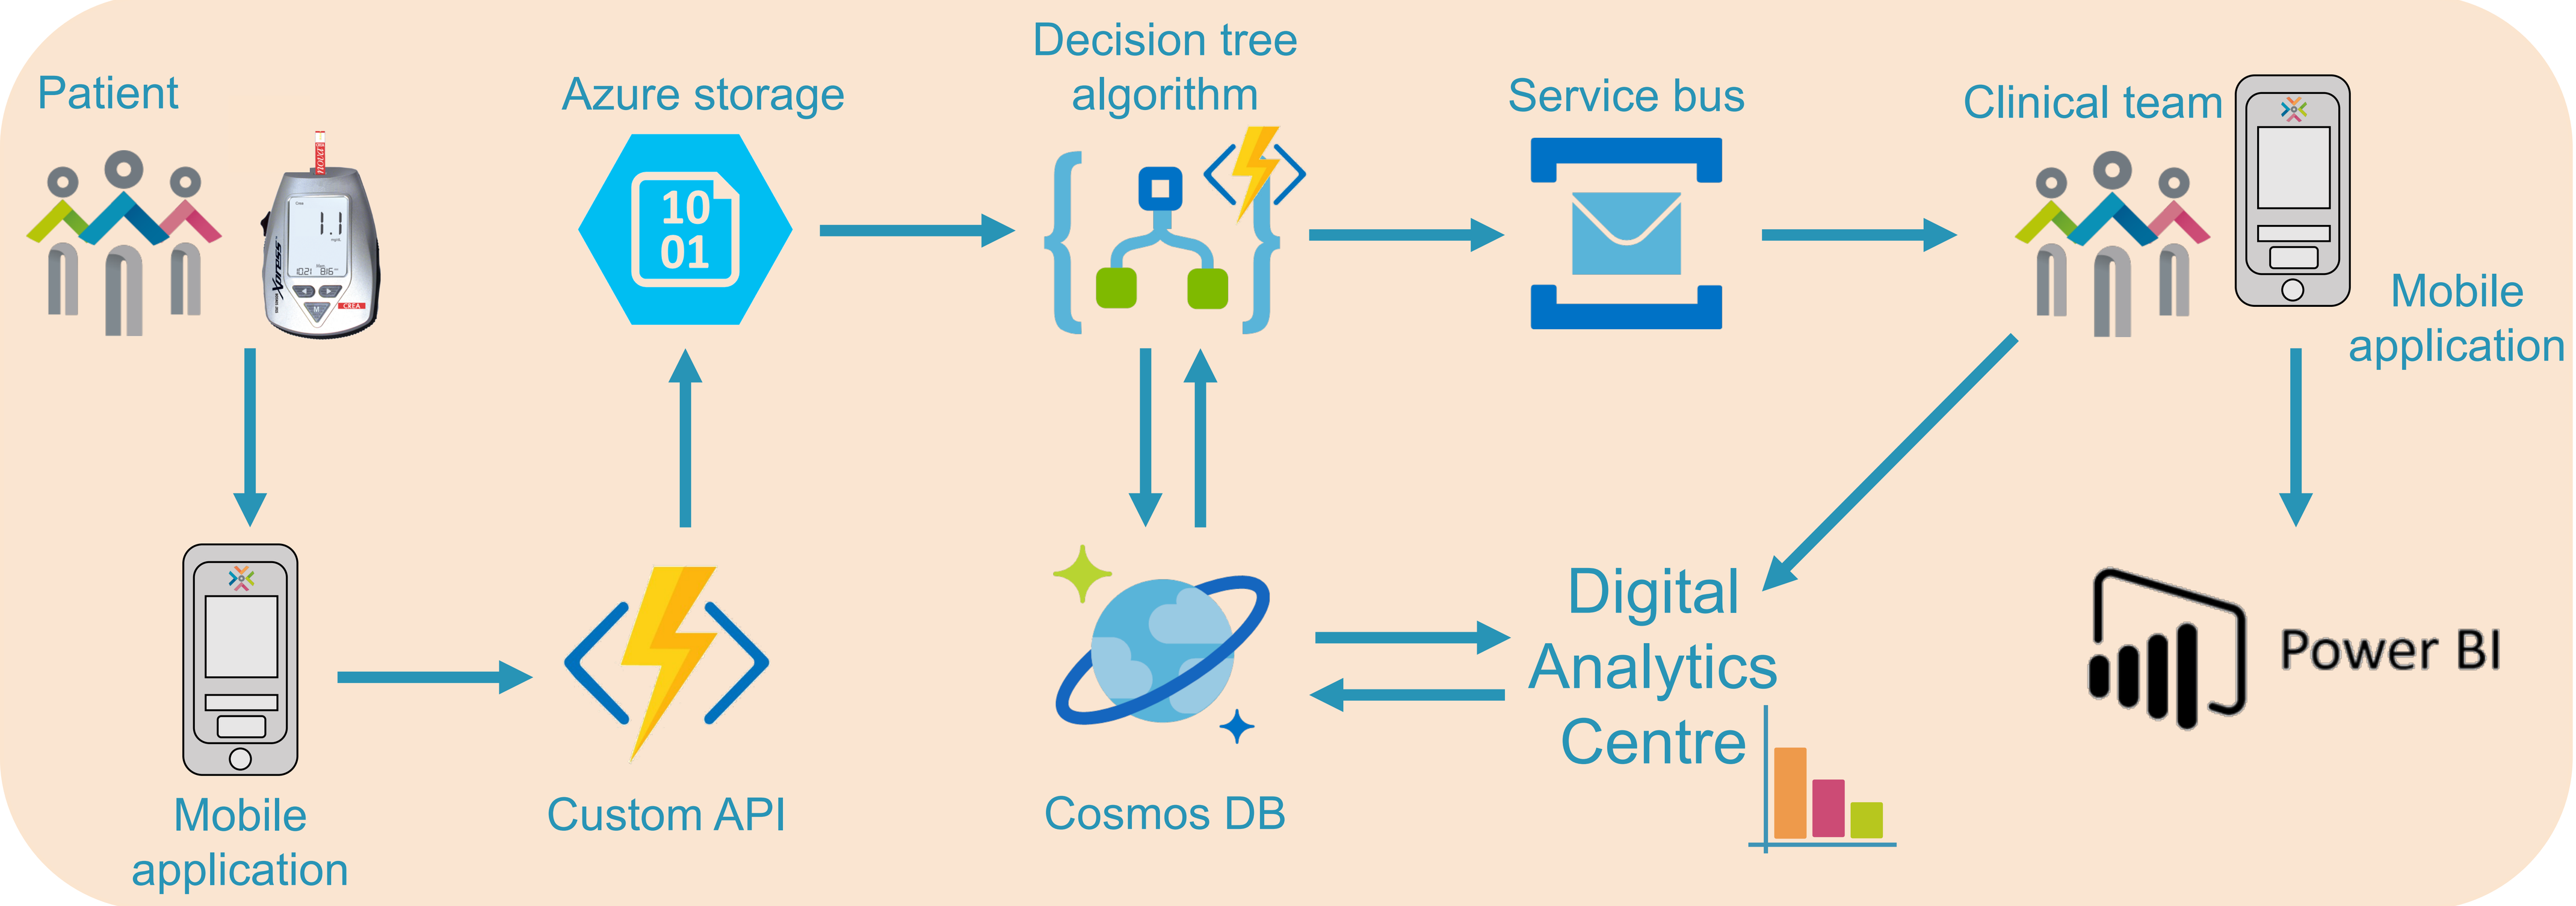
Patient
Azure storage
Decision tree algorithm
Service bus
Clinical team
Mobile application
Mobile application
Custom API
Cosmos DB
Digital Analytics Centre
Power BI

- Secure Microsoft Azure environment
- Data transferred from PowerApps to Azure storage
- End-to-end encryption
- Full audit trail
- Alerts could then be delivered to the clinical team through SMS/email/mobile app

Future development

- Digital clinical trial - scientifically characterise the benefits of digital and technological trial enhancements
- Develop visualisations for patient data
- Develop clinician-facing mobile application
- Connect Microsoft Machine Learning Studio

Acknowledgements
Thank you to Laura Stephenson and Jennifer Bradford for their help and support on this project.

For further information contact
Laura Hutchinson at laura.hutchinson@digitalecmt.org

Engaging patients, Driving decisions

Poster presented at Pharmaceutical R&D Information Systems Management Executive Forum (Paris, May 2018)

MANCHESTER 1824 The University of Manchester
CANCER RESEARCH UK | MANCHESTER INSTITUTE