ABCD

Biobanking for Secondary Use of Clinical Samples and Data

Martin Urban (IT RDM Services), Andreas Köhler (Transl. Medicine + Clin. Pharmacology)

Adcknowlegements: *Alexandre Shin-Iti Okamoto, Andreas Ackermann, Anja Mayer, Anna Arndts, Bernd Bräuninger, Birgit Rubensdoerffer, Birgitta Schieder, Cara Williams, Claus Esmark Larsen, Felix Mader, Florian Gantner, Florian Jaumann, Frank Risse, Gerd Munzert, Hans-Jürgen Lomp, Heinrich von Lips, Irene Lorenz, Jasmin Saric, Jeanette Schlagbauer, Joachim Meyer, Judith Barwig, Jürgen Schindler, Jutta Friese, Karsten Quast, Katja Stalder, Katrin Nivens, Martin Prystaz, Martin Heckenberger, Martin Michel, Michael Osswald, Michael Betke-Hornfeck, Michael Brendel, Michael Geng, Michael Koslowski, Monika Richter, Oliver Schanz, Oliver Blattner, Oliver Kleiner, Peter Seither, Rene Kubiak, Robert Schlotte, Selda Samakoglu, Steven Padula, Sylvia Pratz, Thomas Reith, Torsten Dorm, Ulrich Roth, Wiebke Sauter*

Fit for purpose concept

- Different project intentions (e.g. submission relevant analysis vs research) may require different processes, validation requirements and more or less flexibility
- A fit-for-purpose concept with different categories should be implemented:
 1. Decision making
 Results are generated in a way that allows transfer to authorities (e.g. FDA, EMA) and use as part of submissions
 2. Clinical and Pre-clinical research
 Exploratory analysis, hypothesis generation

Analysis request process

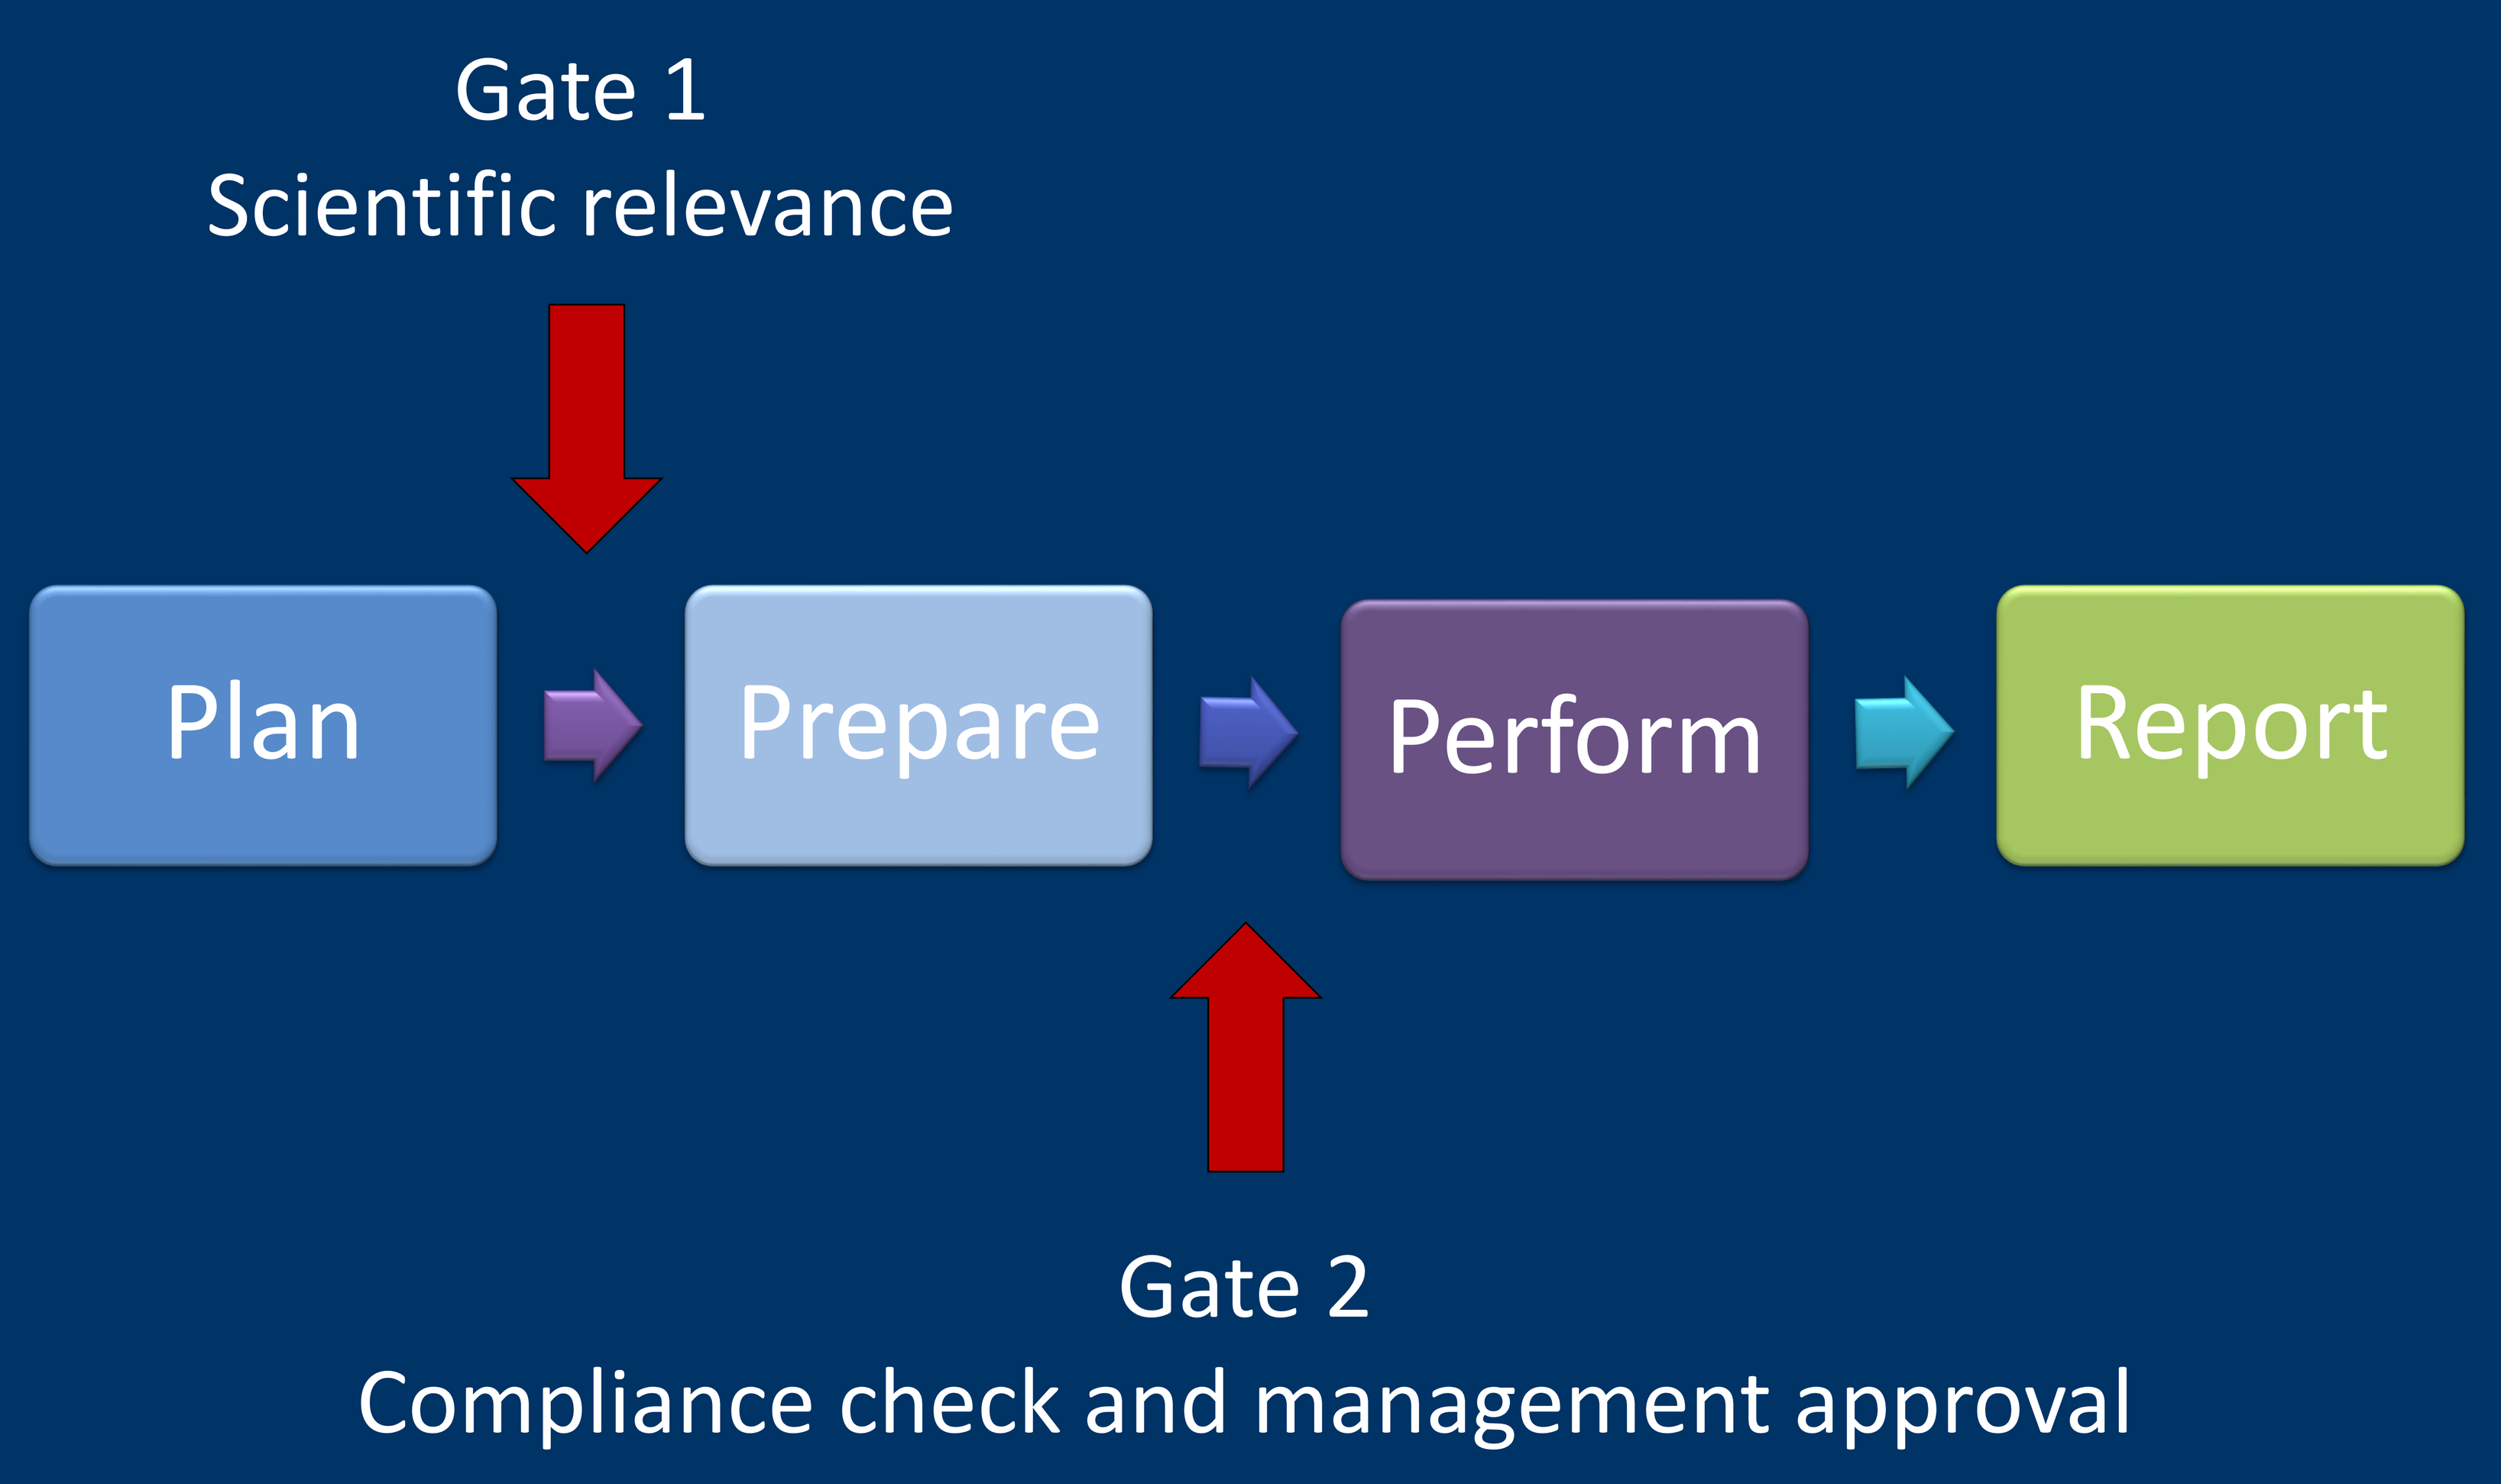
Gate 1
Scientific relevance
Plan
Prepare
Perform
Report
Gate 2
Compliance check and management approval

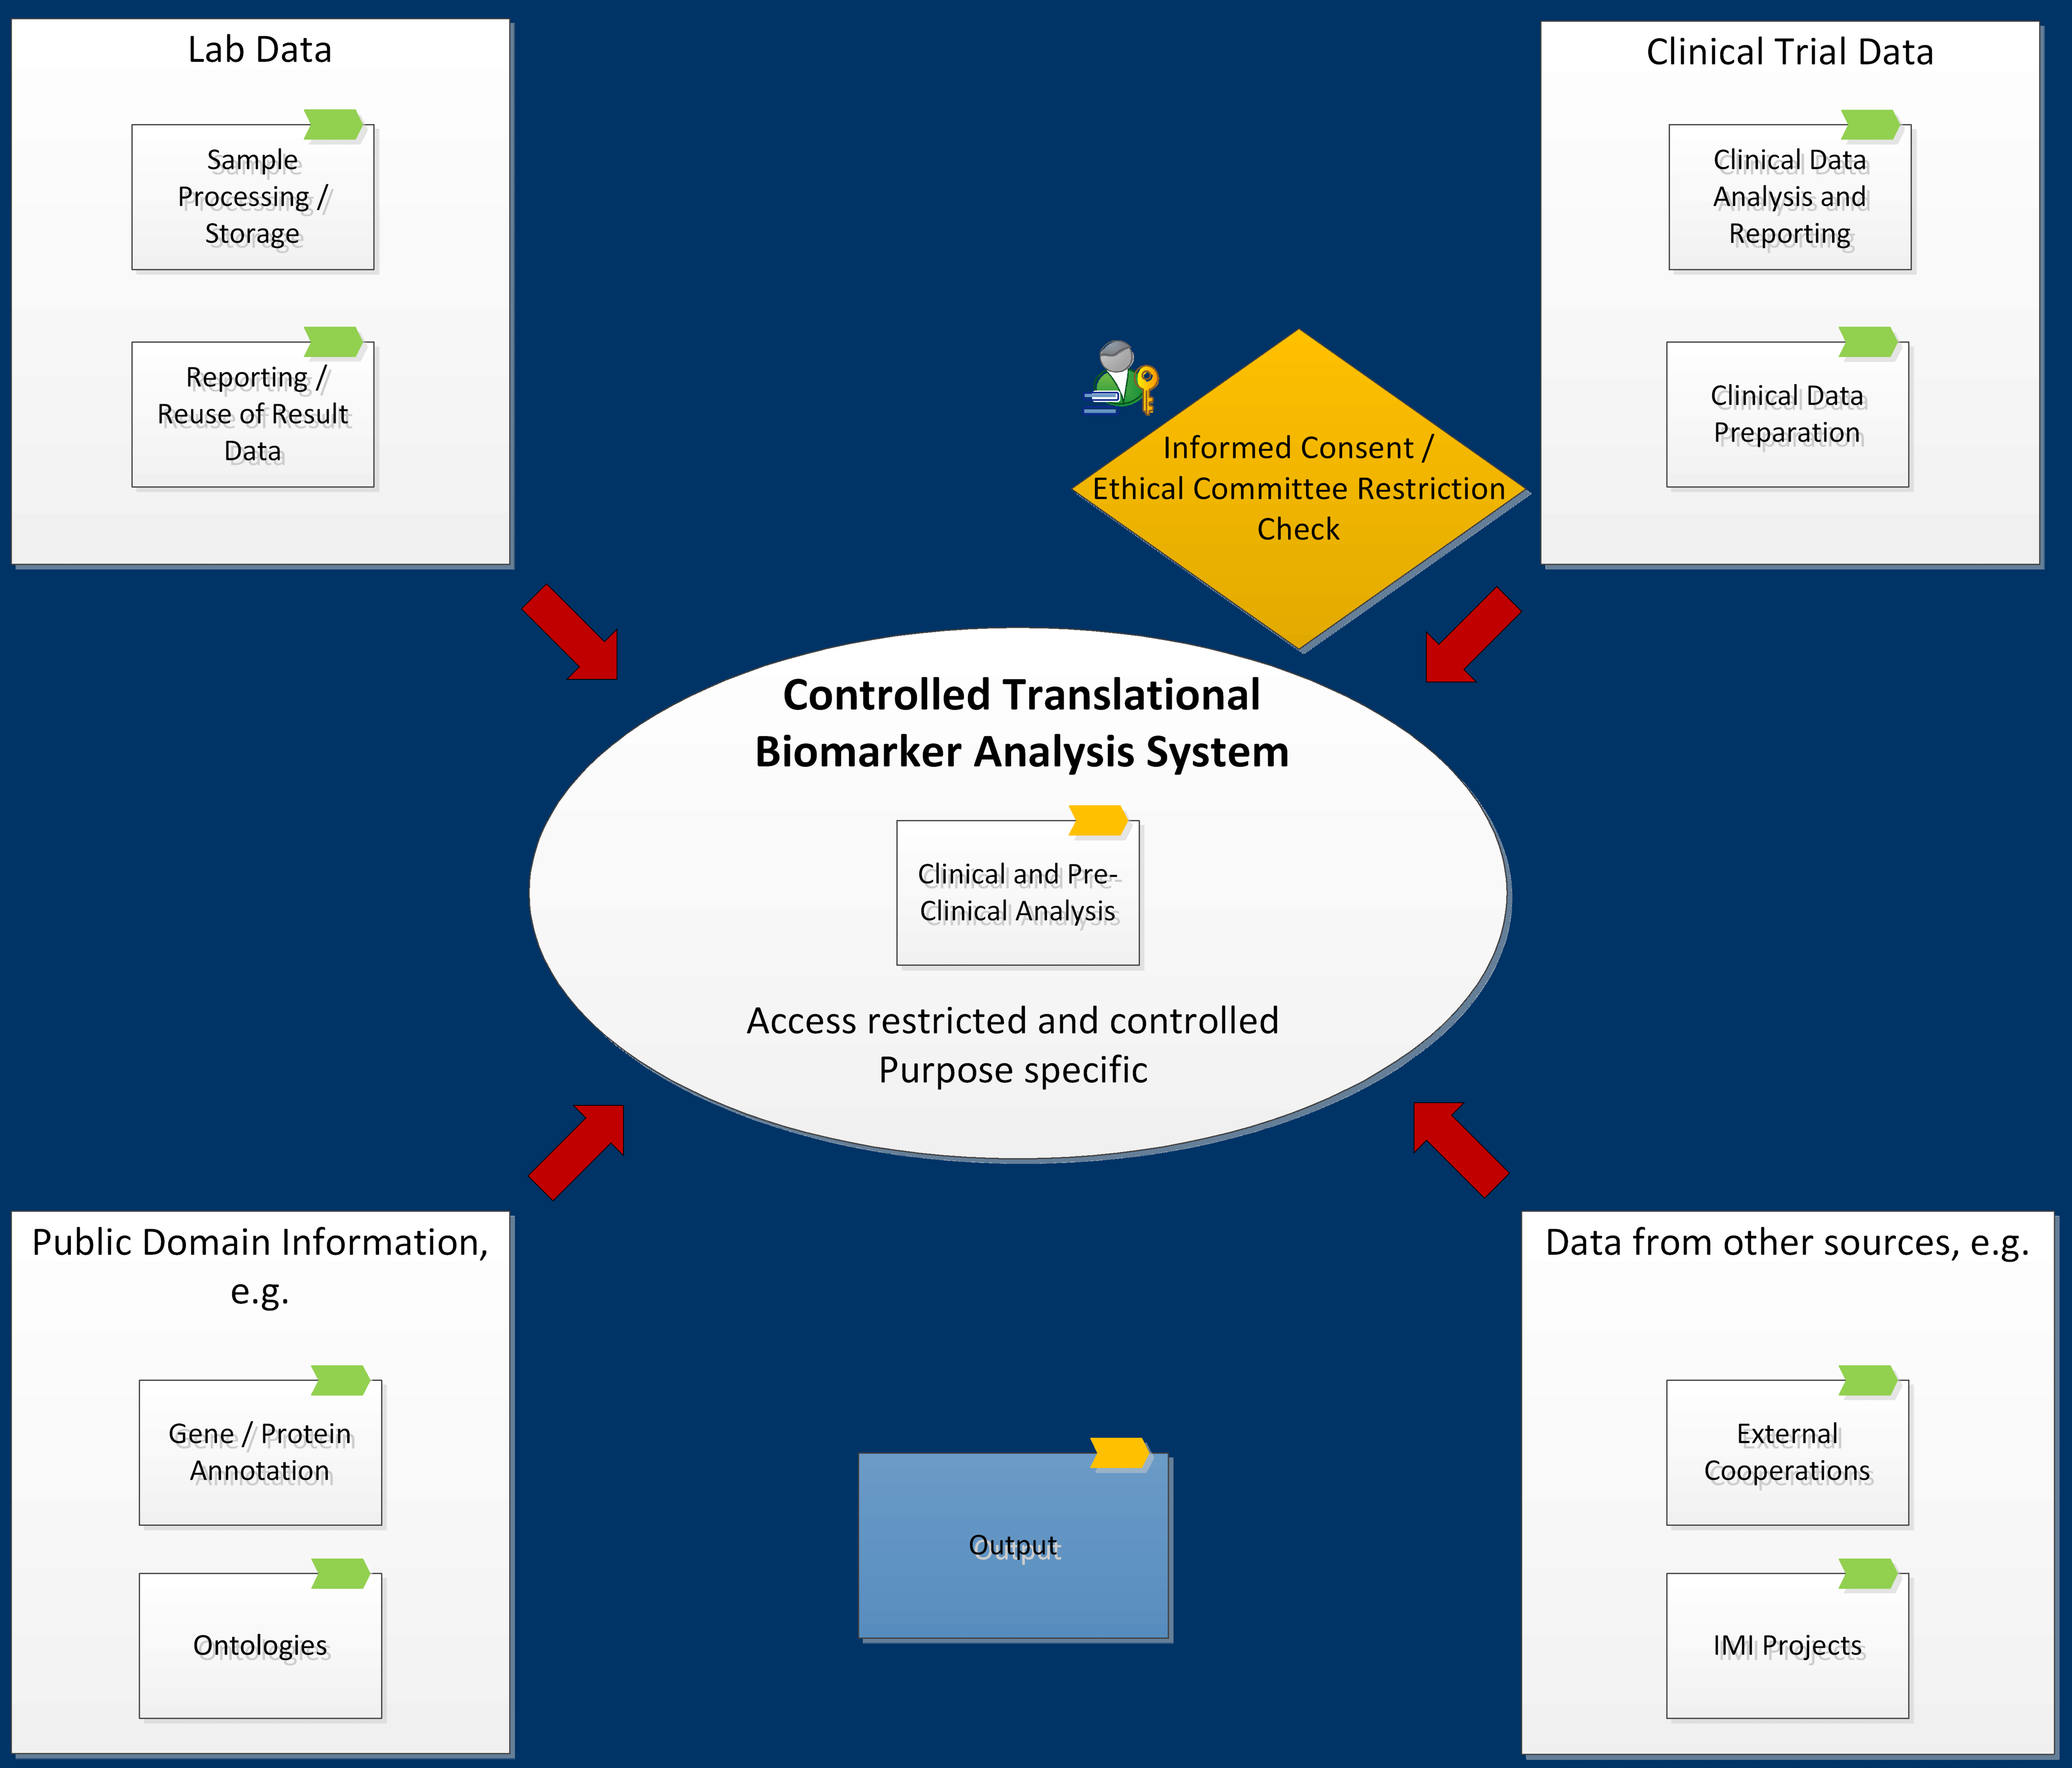
Lab Data
Sample Processing / Storage
Reporting / Reuse of Result Data
Clinical Trial Data
Clinical Data Analysis and Reporting
Clinical Data Preparation
Informed Consent / Ethical Committee Restriction Check
Controlled Translational Biomarker Analysis System
Clinical and Pre-Clinical Analysis
Access restricted and controlled
Purpose specific
Public Domain Information, e.g.
Gene / Protein Annotation
Ontologies
Output
Data from other sources, e.g.
External Cooperations
IMI Projects